A durable gene editing approach for Hemophilia A that enables curative levels of FVIII activity in non-human primates

Metagenomi THERAPEUTICS

Paul Szymanski[1], Janet J. Mei[1], Kalya A. Stanten[1], Junkai Wang[1], Krushangi N. Shah[1], Ashley T. Quintana[1], Allison Quatch[1], Juliette Gafni[1], Bokyoung Song[1], Chia-Yao Lee[1], Luis Gamboa[1], Lana Harshman[1], Snigdha Chatterjee[1], Elaine Chu[1], Keirstinne Turcois[1], Artur Karasyov[1], Wyatt T Clark[1], Steffen Durinck[1], Katalin Kauser[1], and Alan R Brooks[1]

1 - Metagenomi Therapeutics, Emeryville, California, USA.

5959 Horton St, Emeryville, California 94608

Introduction

Hemophilia A is the most common X-linked inherited bleeding disorder and arises from a deficiency of functional factor VIII (FVIII). Exogenous FVIII replacement and FVIII bypassing agents have improved outcomes for patients with this severe disease but have not eliminated breakthrough bleeding and are not curative. Efforts to develop an AAV based FVIII gene therapy have been challenging due to loss of FVIII expression over time[1], and the desire for a cure[2] remains unfulfilled. Gene editing technology may overcome these durability challenges by enabling site specific and permanent integration of a FVIII gene into the genome resulting in persistence of the transgene despite continued cell division.

AIM

We aimed to integrate a B-domain deleted (BDD) FVIII gene in intron 1 of albumin in the genome of hepatocytes under the control of the endogenous albumin promoter to generate stable curative levels of FVIII activity in the plasma after a single *in vivo* treatment.

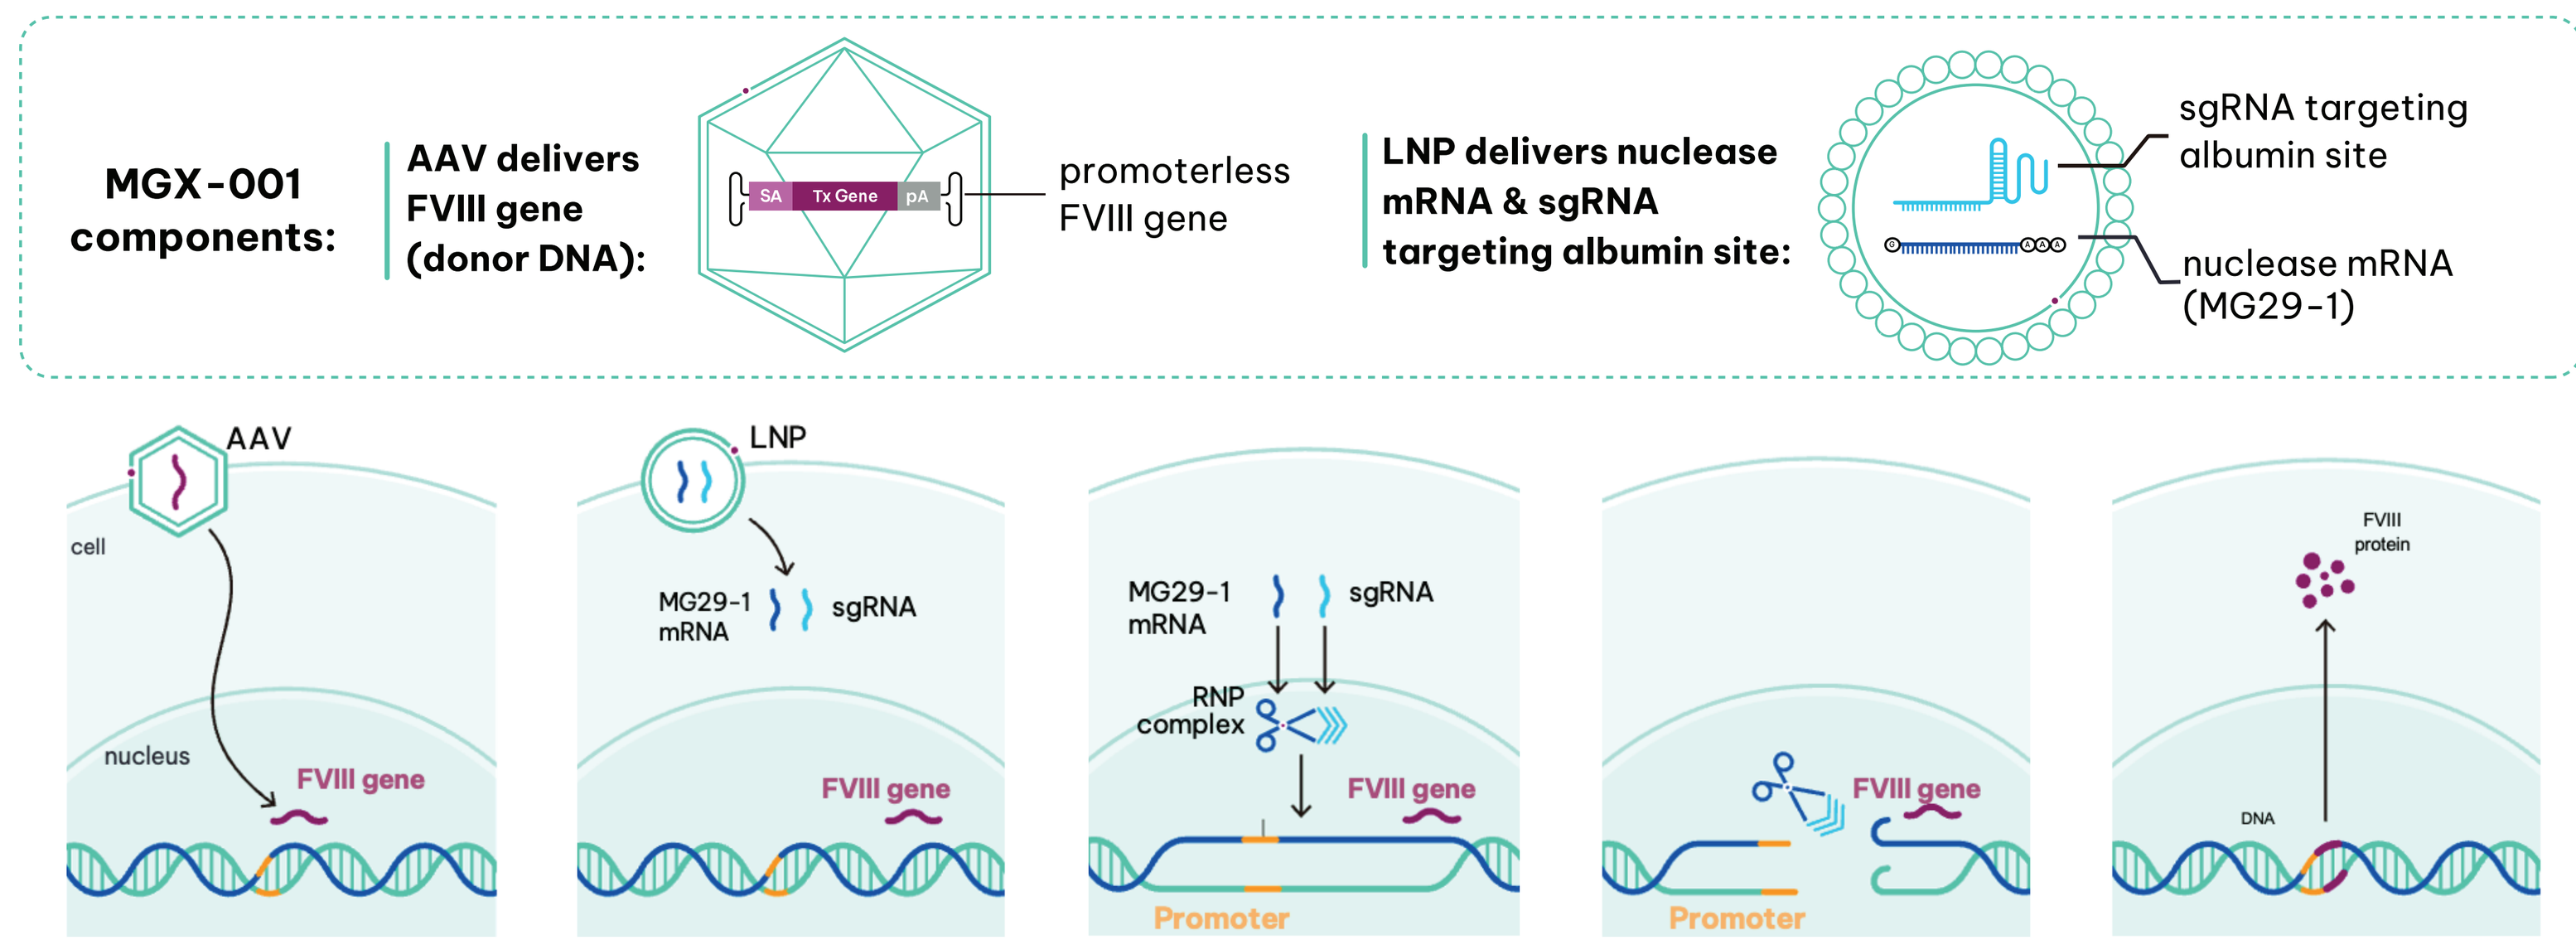
MGX-001 components:
AAV delivers FVIII gene (donor DNA):
SA
Tx Gene
pA
promoterless FVIII gene
LNP delivers nuclease mRNA & sgRNA targeting albumin site:
sgRNA targeting albumin site
nuclease mRNA (MG29-1)
AAV
cell
nucleus
FVIII gene
LNP
MG29-1 mRNA
sgRNA
RNP complex
Promoter
FVIII protein
DNA

AAV delivers promoterless FVIII gene. AAV enters hepatocytes and the donor FVIII cassette localizes to the nucleus.

LNP delivers MG29-1 nuclease mRNA + guide targeting albumin intron 1. mRNA is released in the cytoplasm.

The mRNA is translated into MG29-1 nuclease, which binds the guide RNA to form a ribonucleoprotein (RNP) complex.

MG29-1 nuclease cuts in albumin intron 1, the FVIII gene cassette integrates at the cut site via NHEJ.

The albumin promoter drives the expression of FVIII protein.

Methods

- Non human primates (NHP) were dosed sequentially with two components: a liver tropic AAV delivers a promoter-less FVIII gene cassette and a hepatocyte targeted lipid nanoparticle (LNP) delivers a guide RNA (gRNA) targeted Type V CRISPR nuclease (MG29-1).
- The MG29-1 nuclease makes a double strand break in intron 1 of the albumin gene at which the FVIII gene cassette is inserted by non-homologous end joining (NHEJ).
- Expression of the integrated BDD-FVIII gene is driven from the strong albumin promoter by splicing from albumin exon 1 which adds the albumin signal peptide to the N-terminus of the BDD-FVIII to promote secretion into the circulation.
- To enable evaluation of durability we used the native cyno monkey FVIII (cFVIII) gene sequence that was modified with a single amino acid change which enables measurement of FVIII activity derived from the integrated cFVIII transgene.
- Plasma FVIII activity was monitored in plasma using a chromogenic assay.
- The frequency of FVIII gene integration at the target site was measured in the liver at the end of the study.
- Safety endpoints including clinical chemistries and hematology panels were monitored and histological analysis was performed at study termination.
- Potential sites of off target editing were identified by a combination of in silico prediction, a biochemical method (SITE-seq) and an in cell assay. Potential sites were evaluated by amplicon sequencing of primary human hepatocytes edited at superstaturating doses.

Conclusions

- Site specific integration of the full length hFVIII-BDD gene into the albumin locus in non-human primates (NHP) was achieved using a highly specific type V CRISPR-Cas nuclease.
- hFVIII-BDD was expressed under the control of the endogenous albumin promoter and secreted into circulation.
- This strategy is designed to confer durability that is intrinsically resistant to the episomal loss and transcriptional silencing that are major challenges for AAV gene therapy.
- Plasma levels of hFVIII activity in the normal range were achieved in NHP.
- Plasma FVIII levels were stable for 19 months in NHP (the duration of the study), demonstrating the potential for this approach to provide a life-long cure for Hemophilia A from a single treatment.
- Dose finding studies in NHP identified optimal doses for AAV and LNP that achieved plasma FVIII activity in the normal range at low integration frequencies, a favorable safety profile, no detectable off-target editing and minimal impact on albumin levels.
- These data provide a solid foundation to support progression of MGX-01 into clinical development.

Results

Figure 1: Durability study in NHP.

Three NHPs received an intravenous dose of 2e13 vg/kg of the AAV8-cFVIII viral vector followed 35 days later by 1mg/kg of an unoptimized liver-tropic LNP formulation encapsulating the mRNA encoding the MG29-1 nuclease and albumin targeting gRNA. Panel A: cFVIII activity over 19 months measured using a modified chromogenic assay specific to the transgene encoded cFVIII protein. Panels B through E: A subset of safety markers monitored over the course of the study. Animal 1003 was found dead on day 540 post LNP, autopsy revealed an intussusception of the colon, cause of death was assessed as un-related to the test article.

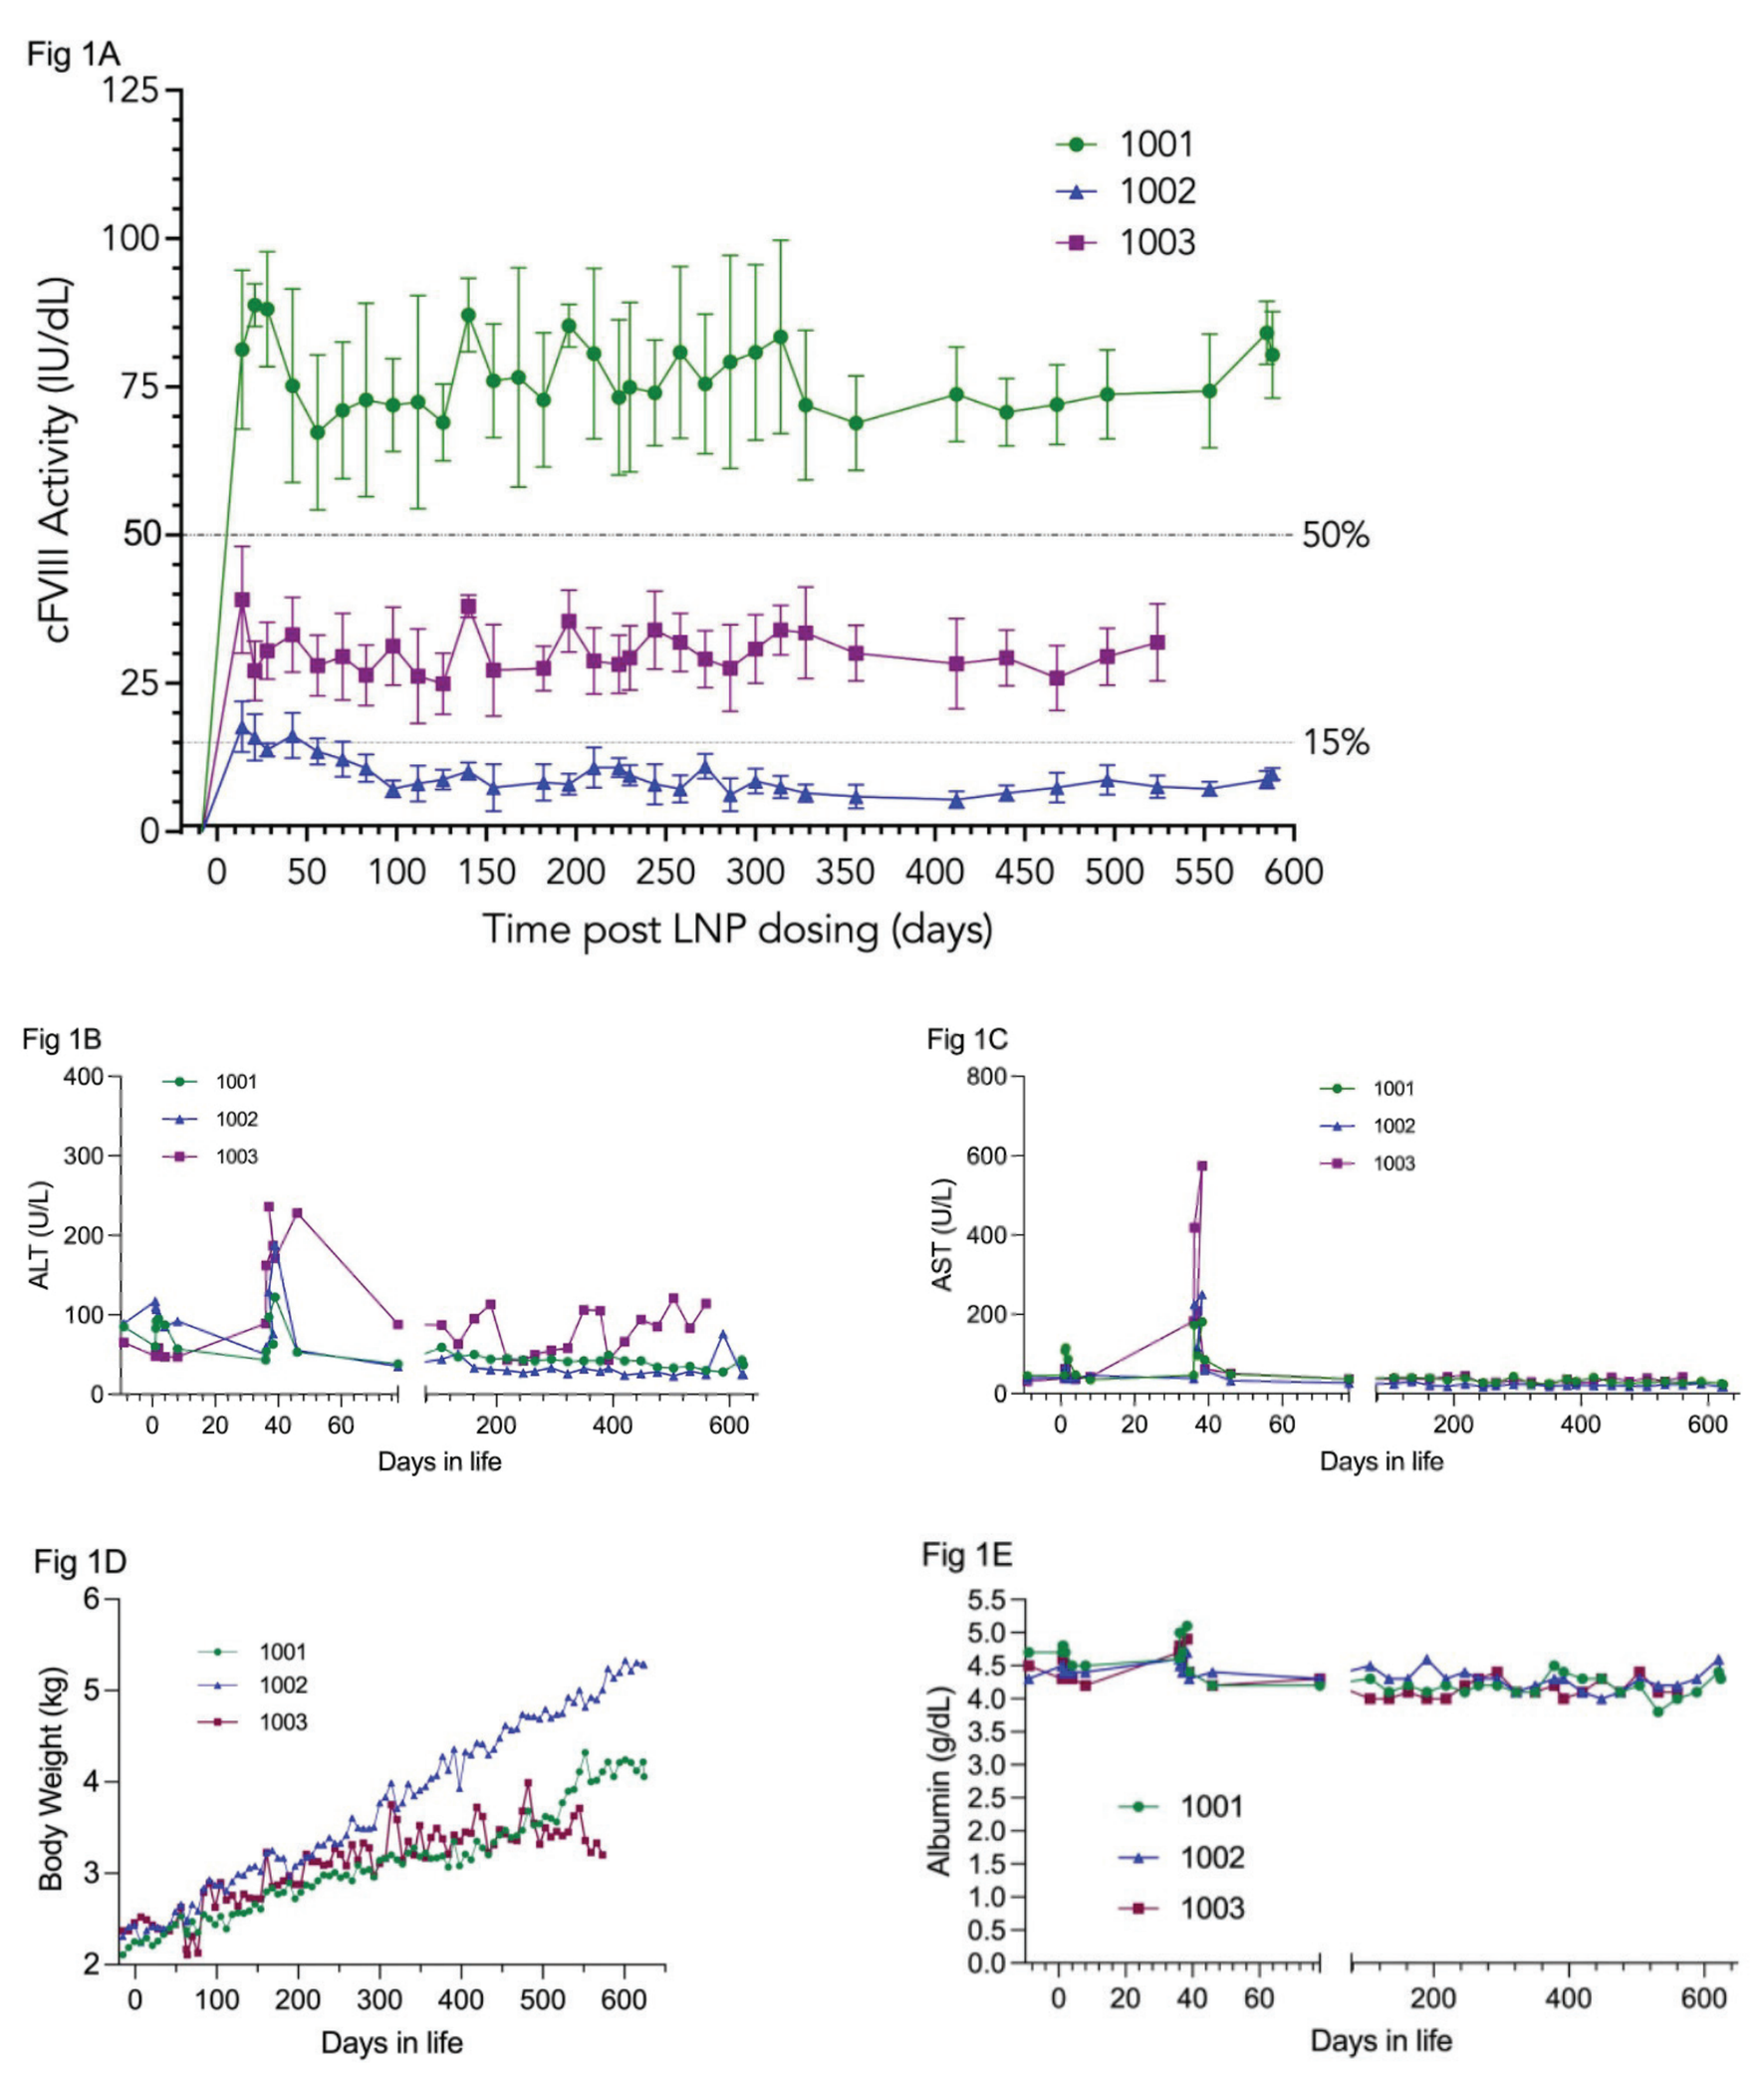
Fig 1A
cFVIII Activity (IU/dL)
Time post LNP dosing (days)
1001
1002
1003
50%
15%
Fig 1B
ALT (U/L)
Fig 1C
AST (U/L)
Days in life
Fig 1D
Body Weight (kg)
Fig 1E
Albumin (g/dL)
Days in life

Figure 2: Dose range finding studies in NHP.

Four NHPs per group received an intravenous dose of the AAV8-hFVIII viral vector followed 21 days later by a liver-tropic LNP formulation encapsulating the mRNA encoding the MG29-1 nuclease and albumin targeting gRNA at different combinations of AAV and LNP doses as shown. hFVIII activity shown is the mean of values on day 5, 8 and 11 post LNP dosing measured using the capture-chromogenic assay. Bars are the mean and standard deviation of each group. A: AAV dose response. B: LNP dose response.

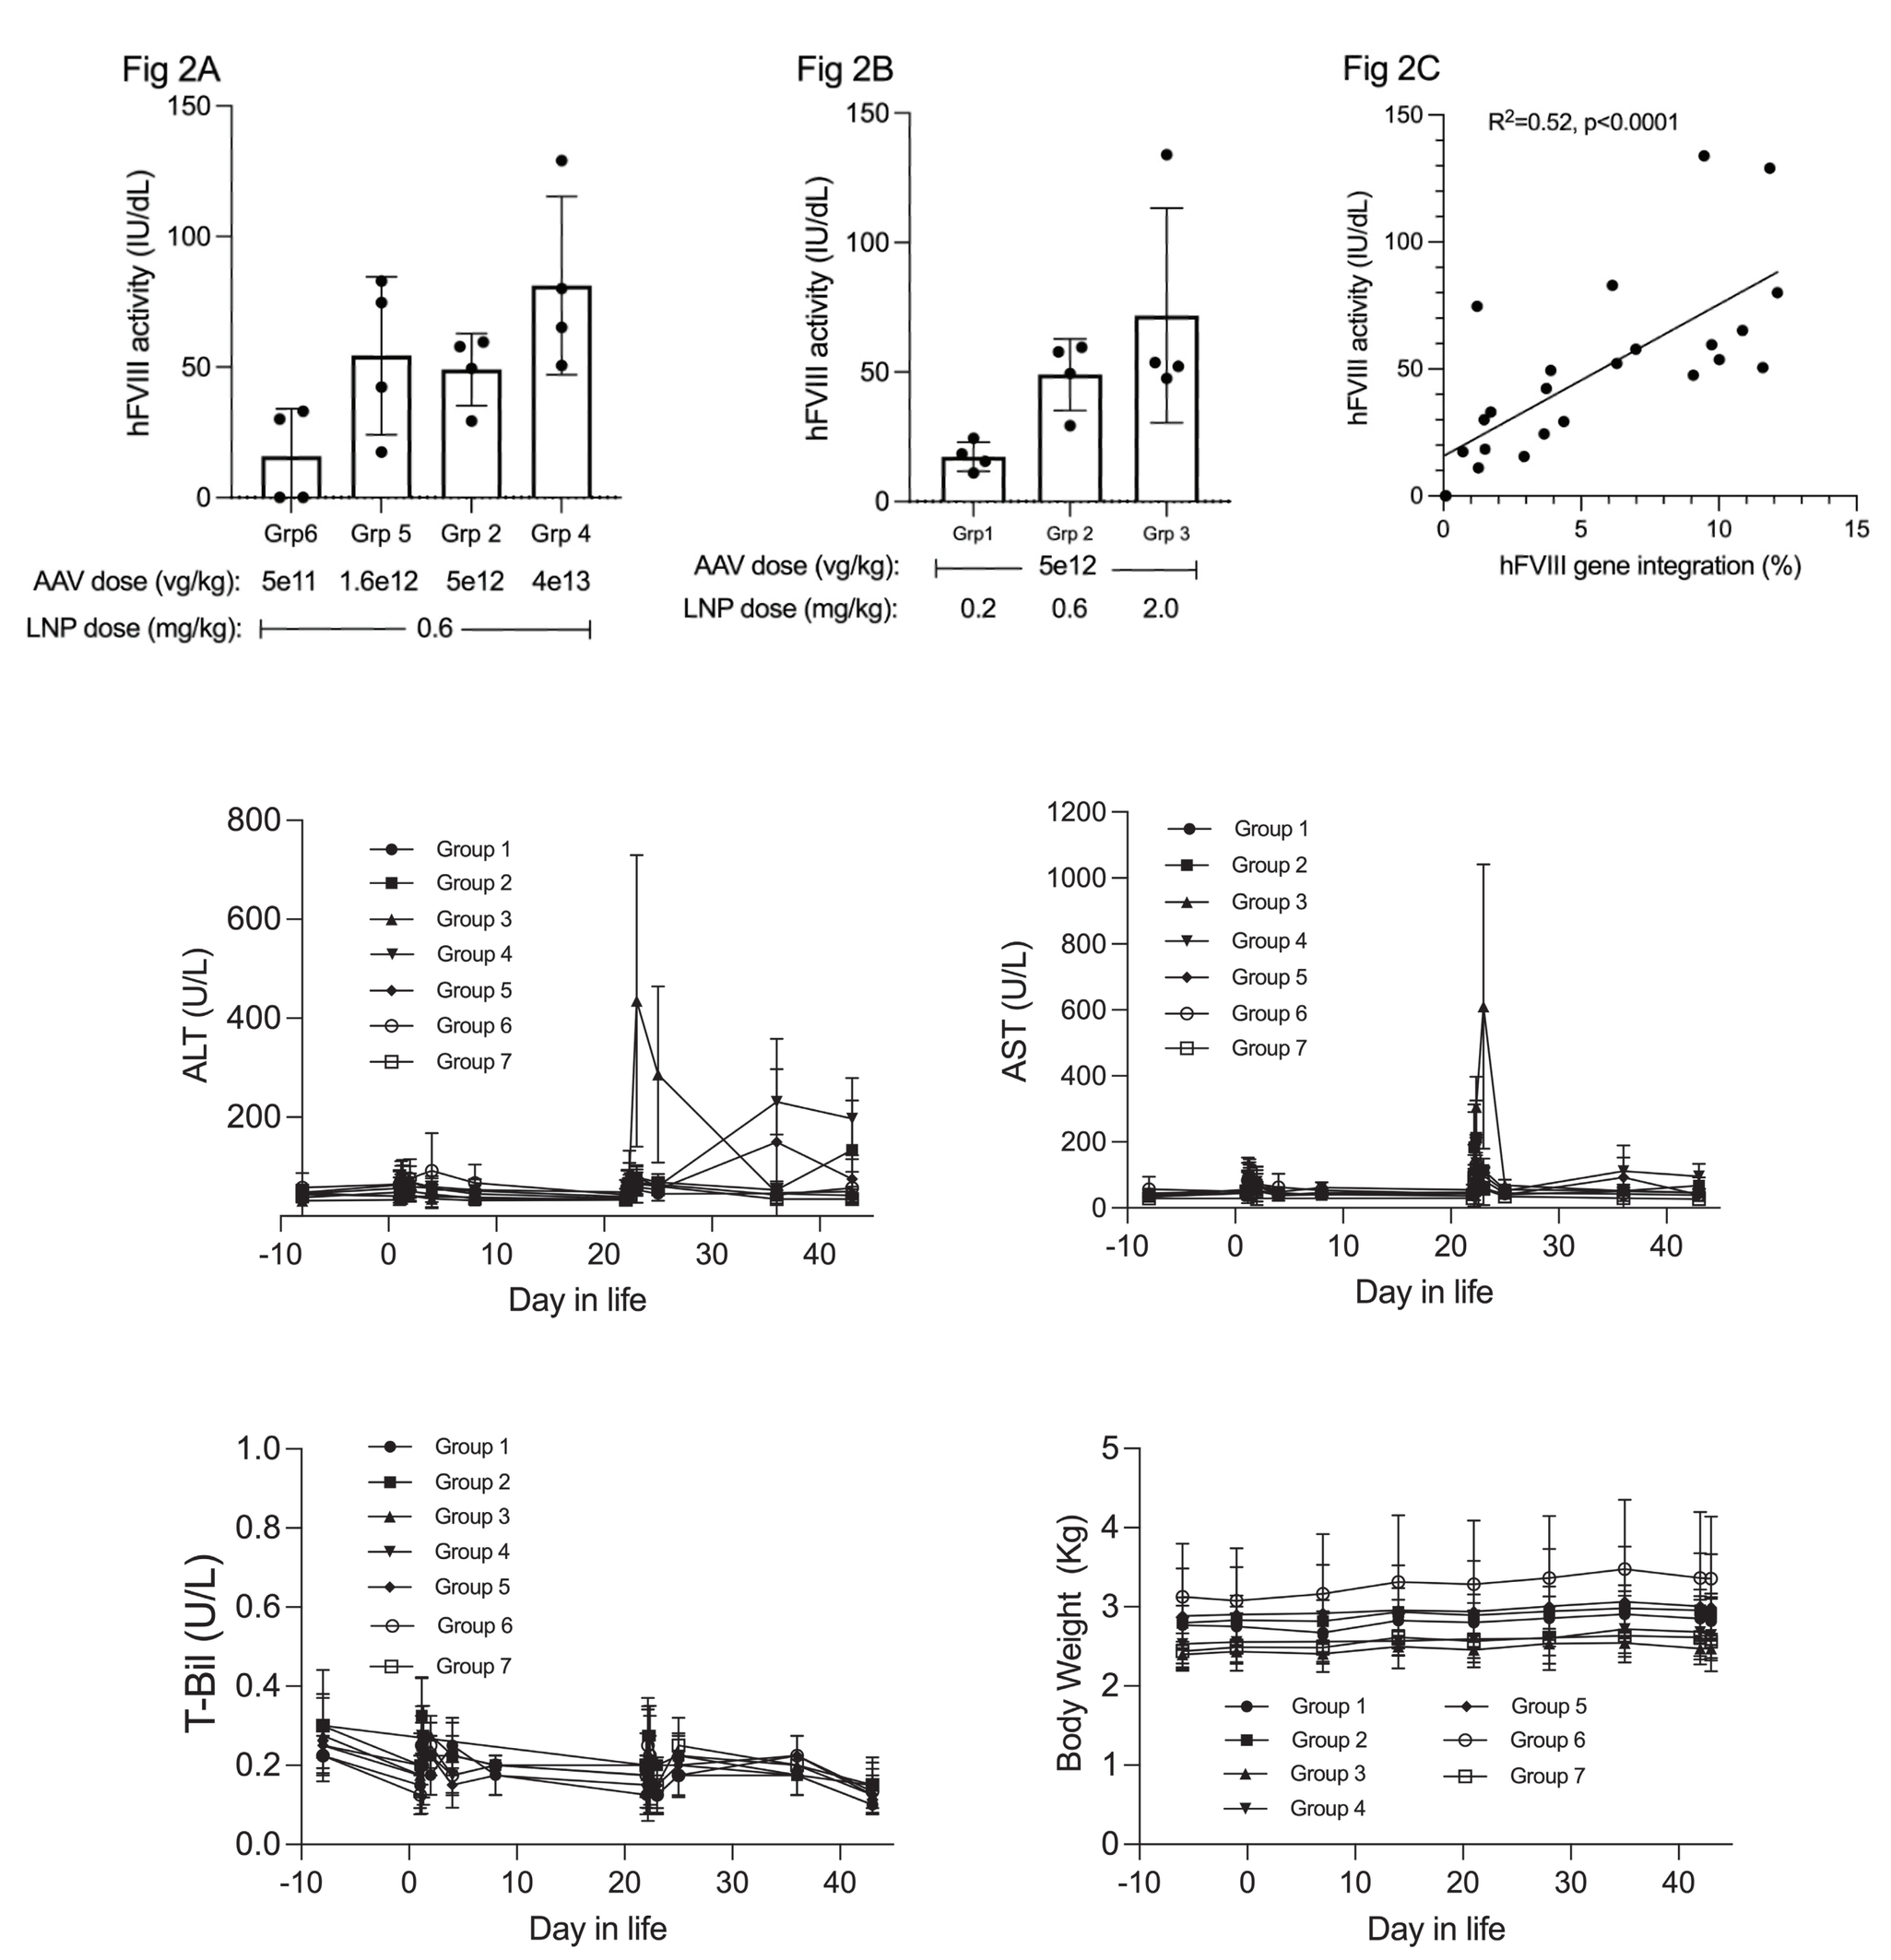
Fig 2A
hFVIII activity (IU/dL)
Grp6
Grp 5
Grp 2
Grp 4
AAV dose (vg/kg): 5e11 1.6e12 5e12 4e13
LNP dose (mg/kg): 0.6
Fig 2B
Grp1
Grp 2
Grp 3
AAV dose (vg/kg): 5e12
LNP dose (mg/kg): 0.2 0.6 2.0
Fig 2C
R²=0.52, p<0.0001
hFVIII gene integration (%)
ALT (U/L)
AST (U/L)
T-Bil (U/L)
Body Weight (Kg)
Day in life
Group 1
Group 2
Group 3
Group 4
Group 5
Group 6
Group 7

Figure 3: Targeted integration and FVIII expression in primary human hepatocytes (PHH).

PHH were transduced with AAV-hFVIII-BDD at a range of MOI followed by exposure to the same LNP used for in vivo studies encapsulating MG29-1 mRNA and gRNA h87 targeting albumin intron 1. Conditioned media from 3 days of culture was assayed for hFVIII activity and gDNA purified from the cells was assayed for integration of the hFVIII gene at the target site in albumin intron 1. Panel A: FVIII activity and integration at a range of AAV doses. Panel B: Correlation between FVIII activity and integration in each replicate culture.

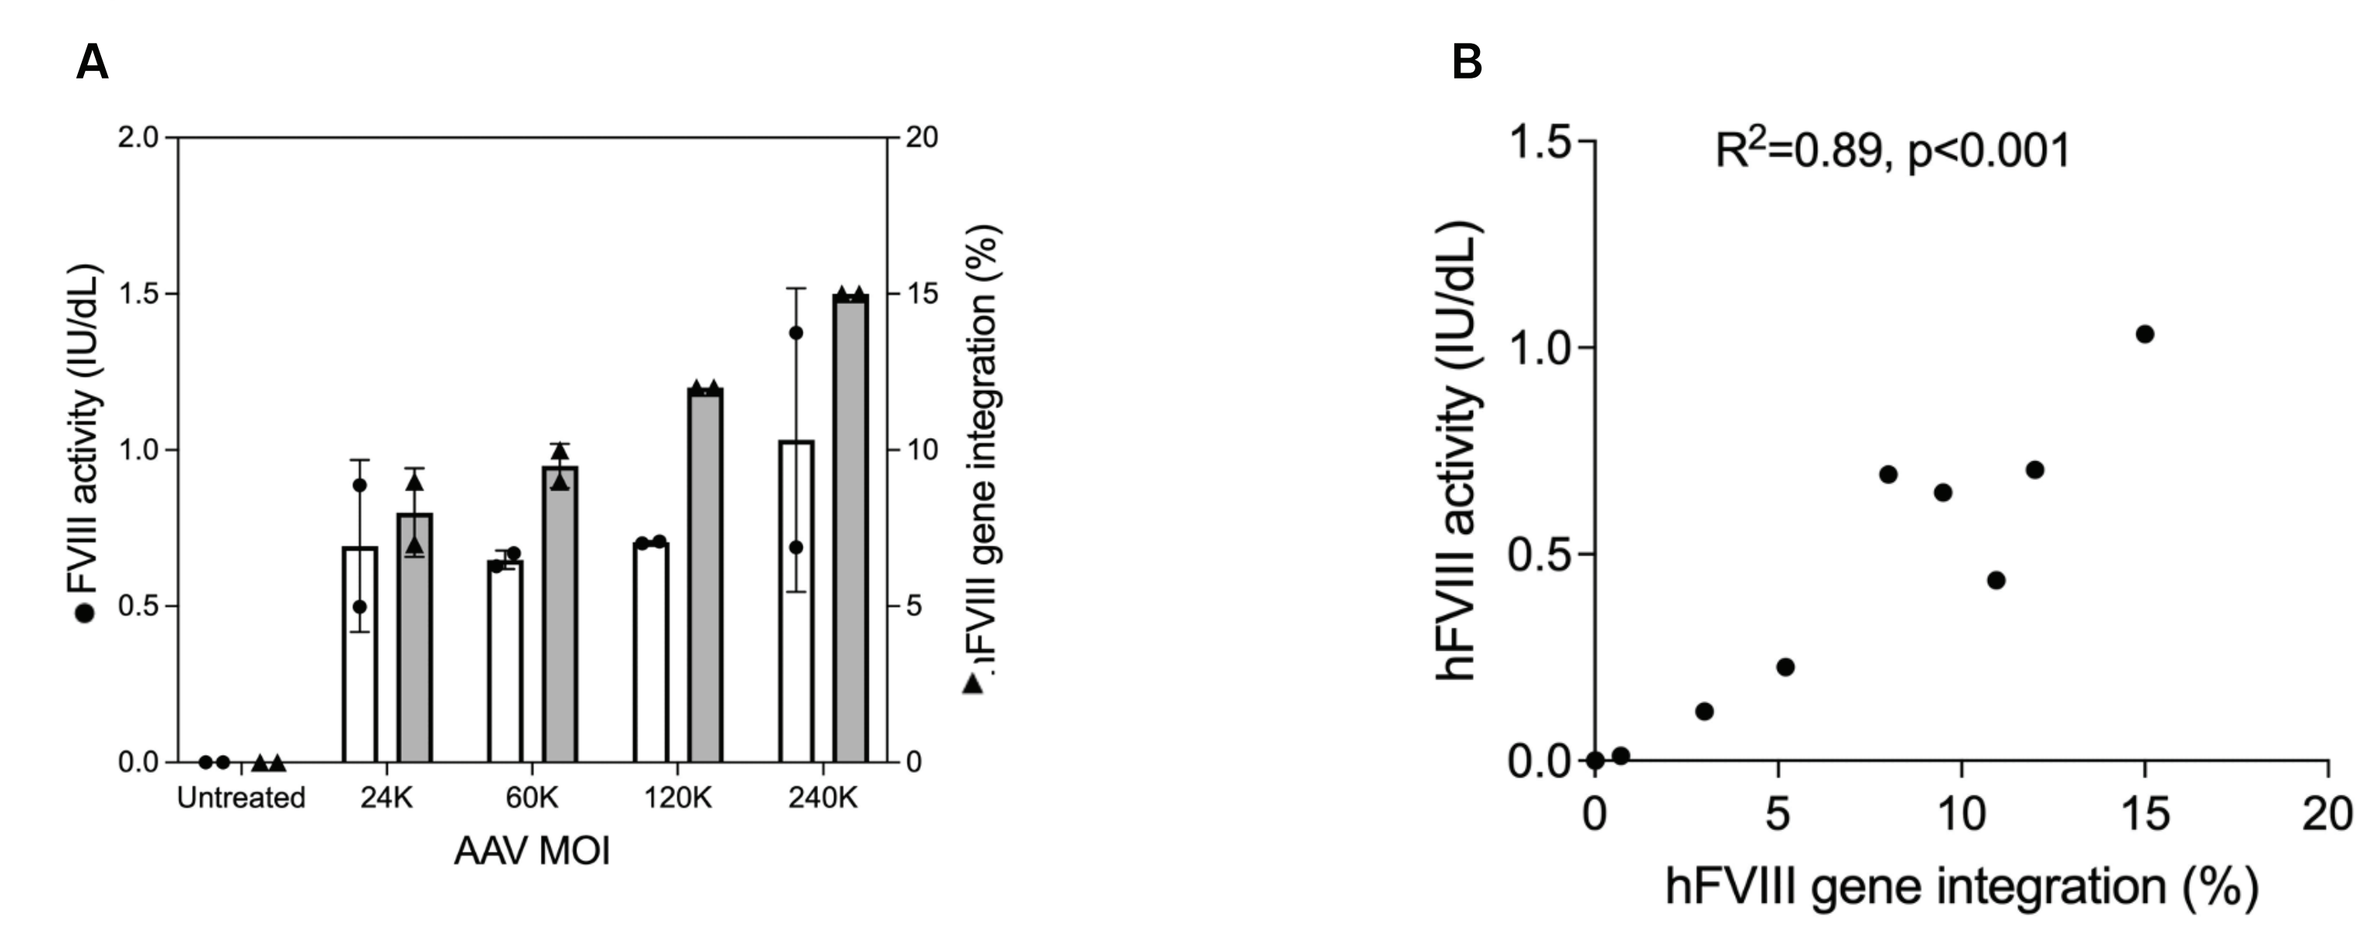
A
FVIII activity (IU/dL)
FVIII gene integration (%)
Untreated
24K
60K
120K
240K
AAV MOI
B
R²=0.89, p<0.001
hFVIII activity (IU/dL)
hFVIII gene integration (%)

Figure 4: Evaluation of off-target (OT) editing.

Panel A: A total of 623 potential OT sites were identified using three orthogonal methods: In silico prediction using the variant aware CRISPRme algorithm, a biochemical in vitro cleavage assay (SITE-seq) and an in cell method (GUIDE-seq). Panel B: Evaluation of 620 of the potential OT sites in PHH edited with LNP delivered MG29-1 mRNA and gRNA h87 at doses resulting maximal editing (1x) as well as doses 8-fold and 32-fold higher. Data are plotted as the editing efficiency (InDel%) in treated (edited) compared to unedited control cells. No sites met the criteria for a validated OT site of having a log-fold-change (LFC) value greater than 1, an average indel rate above 0.1% in treated samples, and a p-value less than 0.05.

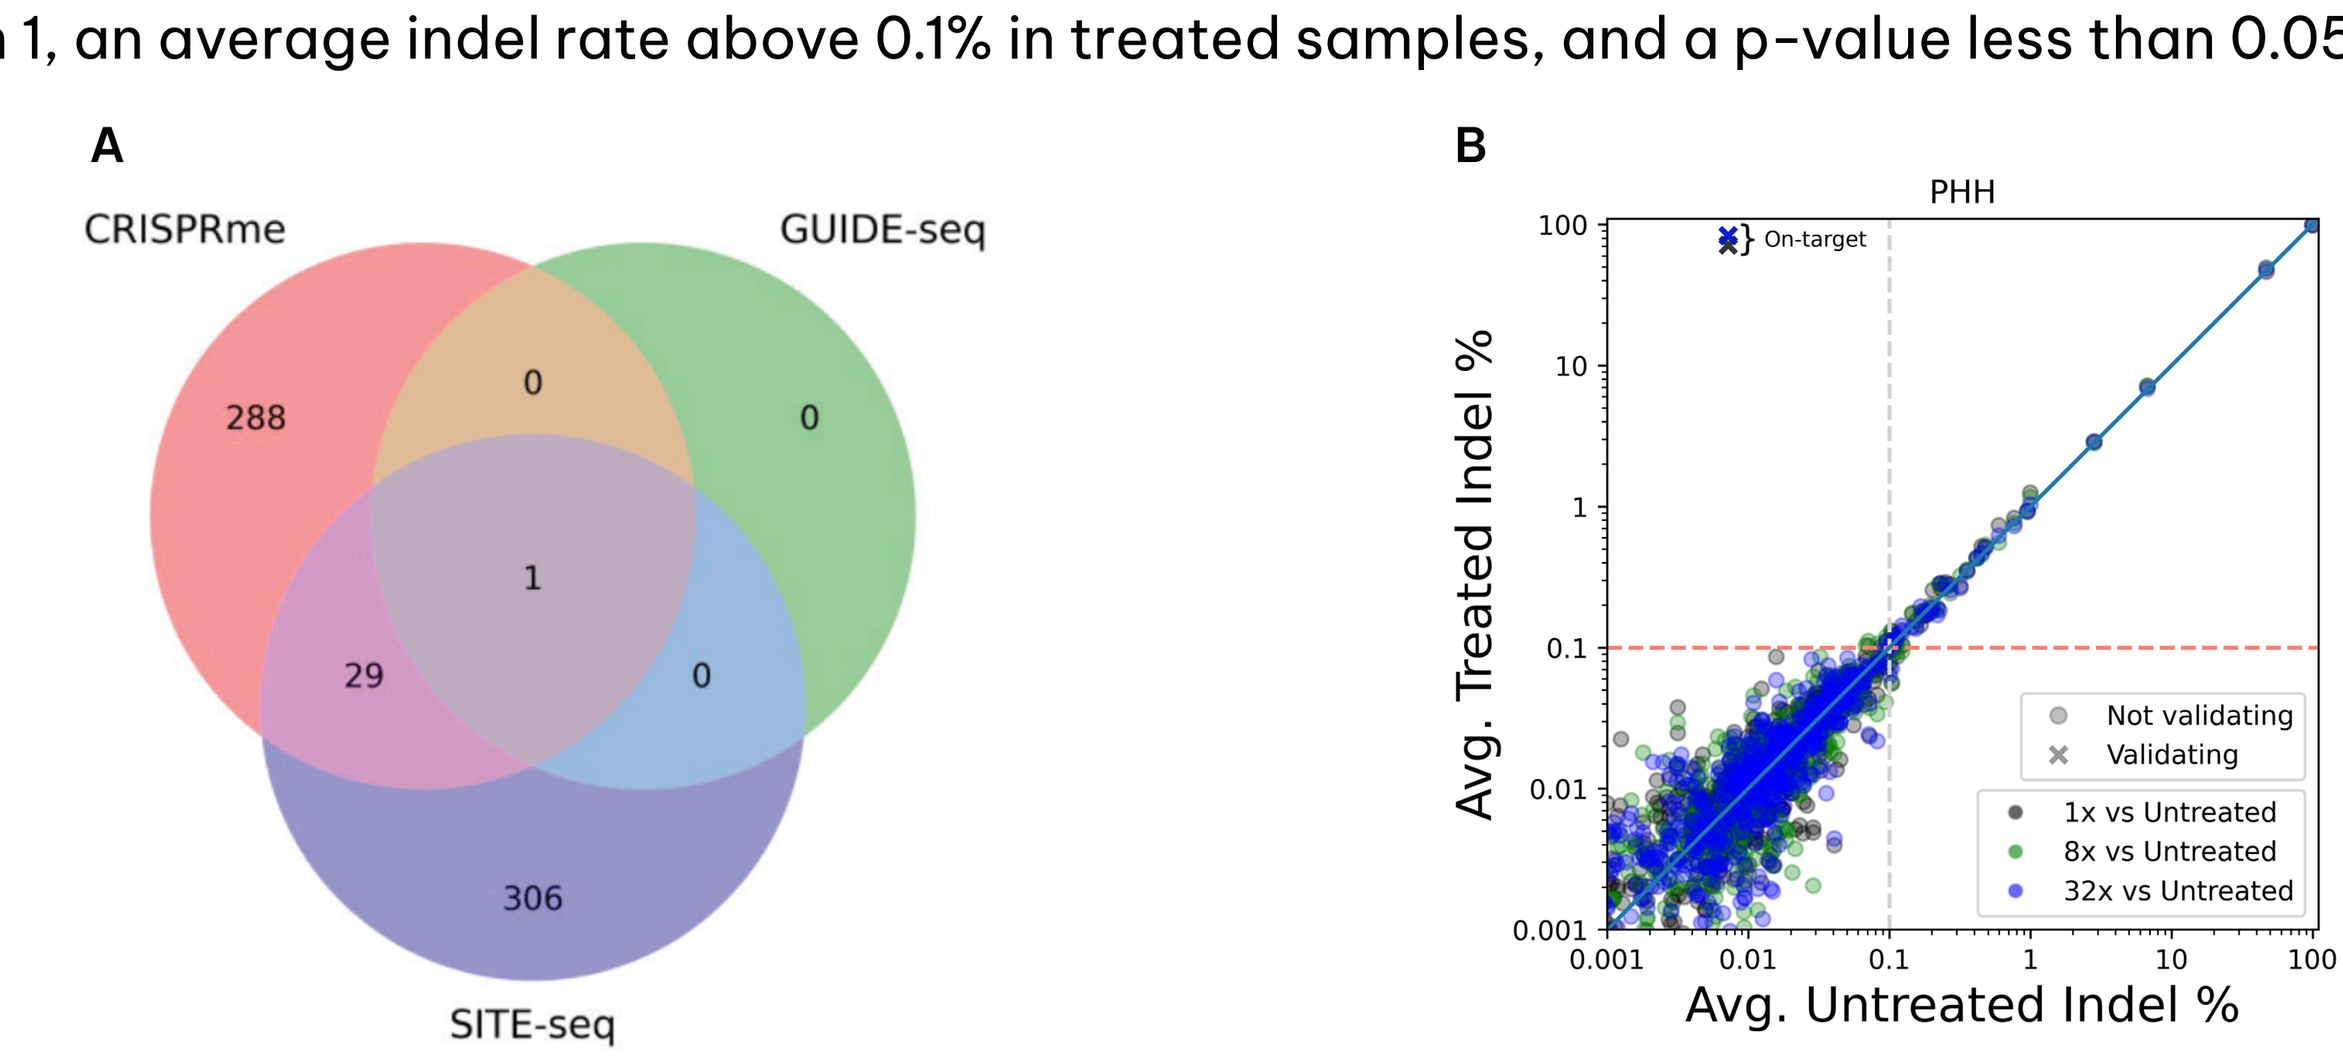
A
CRISPRme
GUIDE-seq
SITE-seq
288
0
0
1
29
0
306
B
PHH
On-target
Avg. Treated Indel %
Avg. Untreated Indel %
Not validating
Validating
1x vs Untreated
8x vs Untreated
32x vs Untreated

Acknowledgements

All NGS was performed by Jake McCurdy and Rachel Szymanski, Jennifer Oki created analysis pipelines for sequence analysis.

References

1. Leavitt A.D. et al. Durability of efficacy, safety, and quality of life 5 years after valoctocogen roxaparvovec gene transfer for severe hemophilia A: final phase 3 GENEr8-1 trial results. *Res Pract Thromb Haemost. 2026: 10:e103416.*
2. Pierce G.F. et al. Why is the uptake of gene therapy for Hemophilia less than expected? *Res Pract Thromb Haemost. 2025: e102948.*

Contact information

Emily Thompson,
Metagenomi Therapeutics 5959 Horton Street, Emeryville, CA99806.
email: emily@metagenomi.co

5959 Horton St,
Emeryville, California 94608
+1 (510) 871-4880
www.metagenomi.co