Stable expression of ATIII in non-human primates by CRISPR-mediated gene knock-in for the treatment of inherited thrombophilias

K. Senger[1], B.M. Justice[1], B. Anggito[1], H. Kalluri[1], B. Pekarek[1], K.M. Stumpf[1], A.A. Quach[1], B. Son[1], L.A. Gamboa[1], S.M. Laperriere1, D. Sim[1], K. Kauser[1], B.C. Thomas[1], P. Szymanski[1], and A.R. Brooks[1]

1 - Metagenomi Therapeutics, Emeryville, California, USA.

5959 Horton St, Emeryville, California 94608

Introduction

Autosomal dominant mutations in the gene encoding ATIII, SERPINC1, result in hereditary thrombophilias with a high risk of serious clotting events[1]. Current standard of care requires lifelong treatment with direct oral anticoagulants (DOACs), which cannot be used during pregnancy[2] and surgery. In these circumstances, antithrombin concentrates are administered. A gene therapy approach would eliminate the need for these treatments and could provide patients with a disease-free mindset. AAV-based delivery of promoter-transgene cargos to the liver has been shown to result in sustained transgene expression in the clinic. However, the non-integrative nature of these therapies can result in the loss of episomal AAVs over time. Our stable genome insertion approach avoids these pitfalls by providing durable, long-term protein expression from the endogenous albumin promoter.

Platform: NHEJ-mediated gene integration at albumin intron 1 converts liver into therapeutic protein "bioreactor"

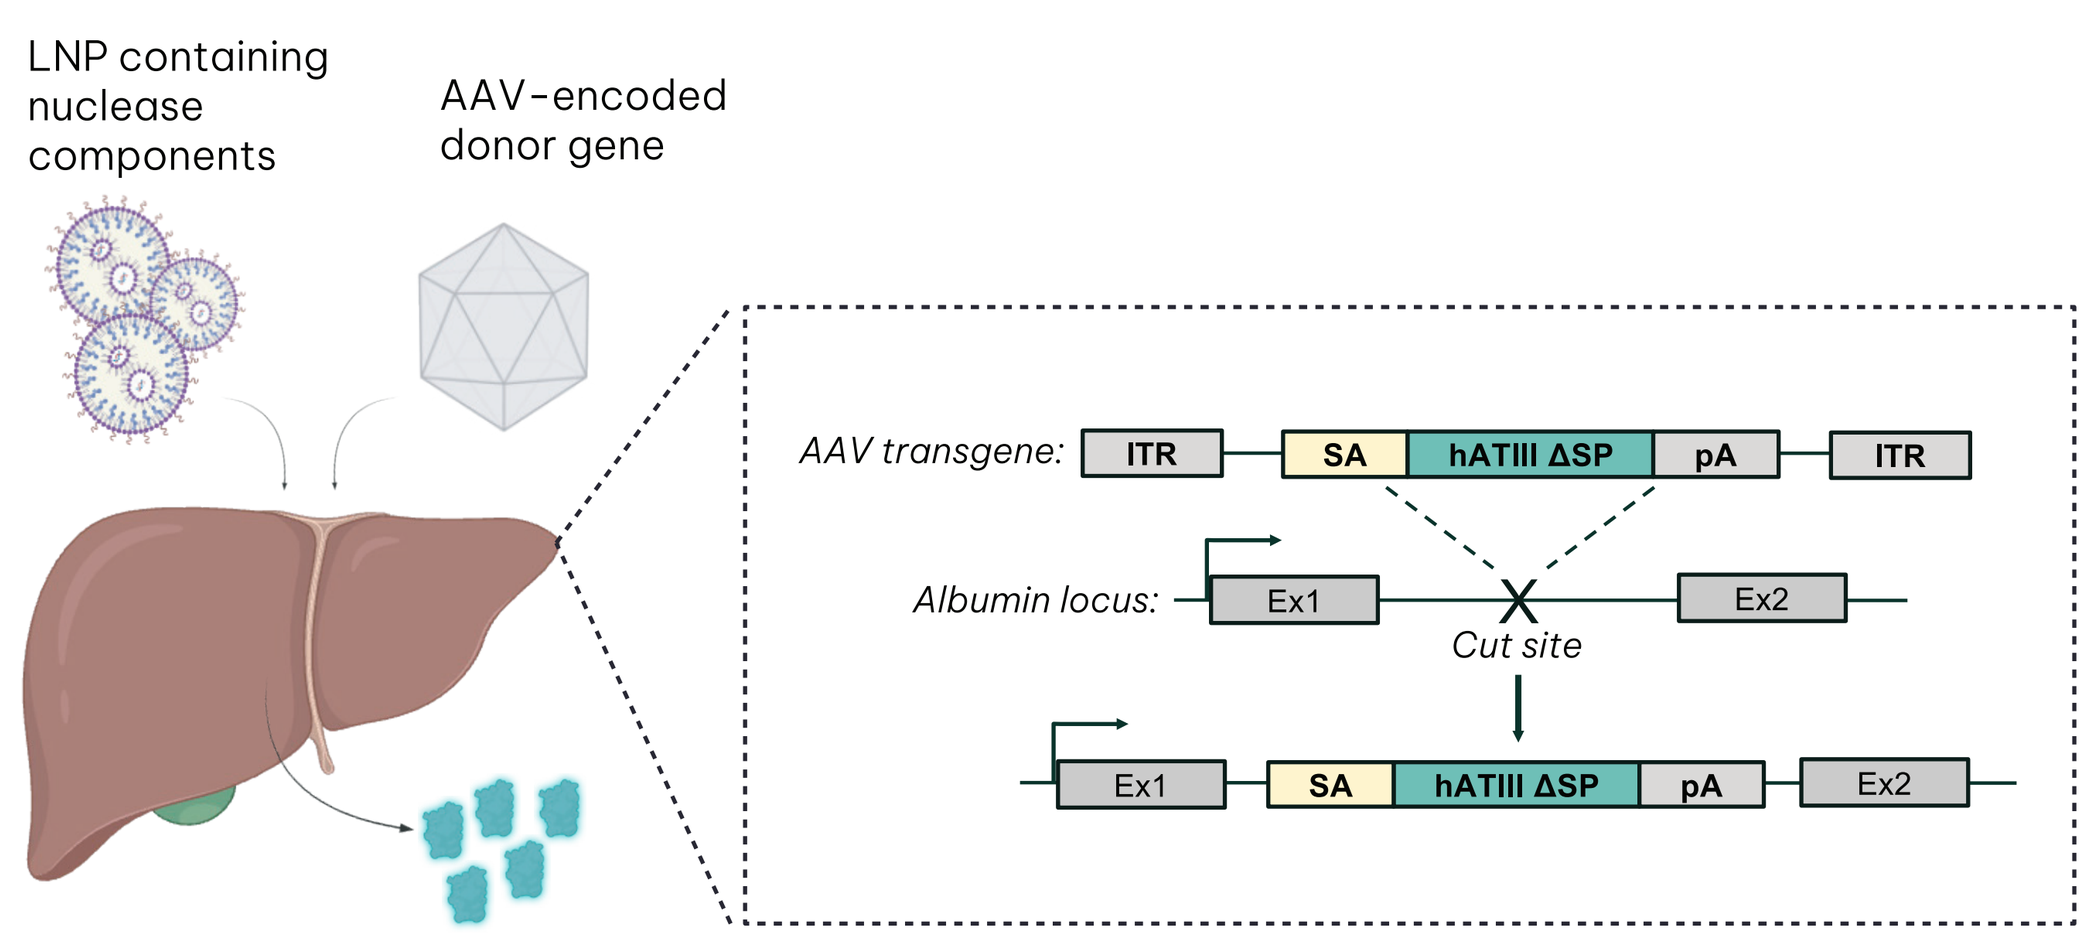
LNP containing nuclease components
AAV-encoded donor gene
Therapeutic protein
AAV transgene:
ITR
SA
hATIII ΔSP
pA
ITR
Albumin locus:
Ex1
Ex2
Cut site

ITR = Inverted terminal repeat, SA = splice acceptor, SP = signal peptide, pA = poly-A

AIM

To achieve stable, therapeutic levels of human ATIII in mice and non-human primates through the permanent insertion of the SERPINC1 gene into the hepatocyte genome using a CRISPR nuclease and AAV-delivered donor gene template.

Method

We delivered three distinct AAV designs encoding the human *SERPINC1* gene in C57Bl/6 mice, followed by a liver tropic LNP to deliver a mRNA encoding a type V CRISPR nuclease and its cognate guide RNA targeting albumin exon 1.
The *SERPINC1* gene from the AAV vector integrates at the albumin cut site, placing *SERPINC1* under the control of the endogenous albumin promoter. We evaluated rates of genome cleavage, transgene integration efficiency, ATIII gene expression, and ATIII biological activity.
A design was selected based on the results in mice for testing in non-human primates.

Results

AAV downselection in mice

AAV template designs tested

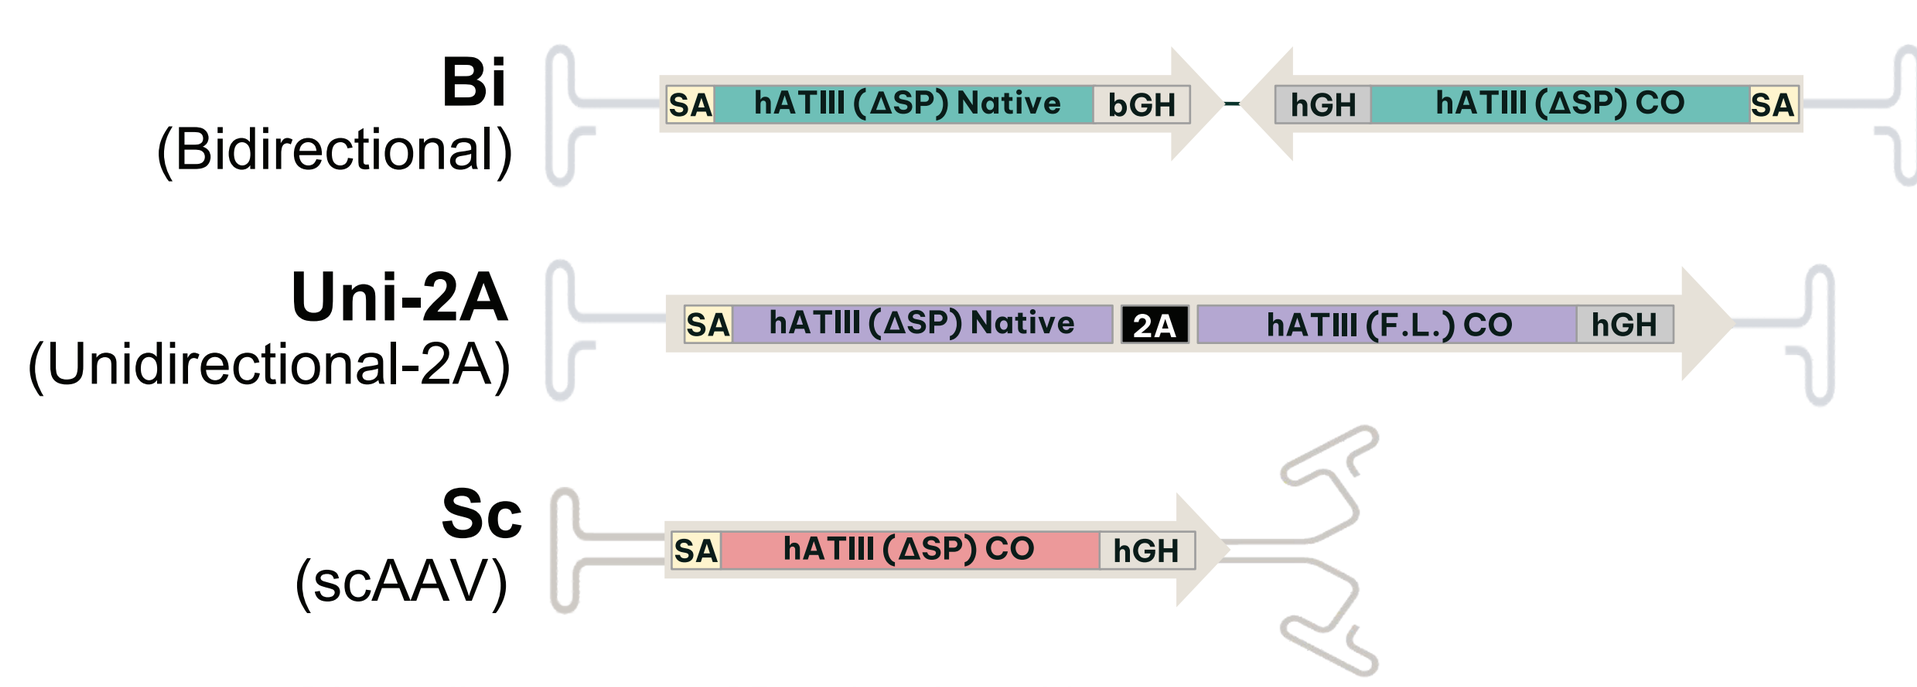
Bi
(Bidirectional)
SA hATIII (ΔSP) Native bGH
hGH hATIII (ΔSP) CO SA
Uni-2A
(Unidirectional-2A)
SA hATIII (ΔSP) Native 2A hATIII (F.L.) CO hGH
Sc
(scAAV)
SA hATIII (ΔSP) CO hGH

SA = splice acceptor; SP = signal peptide; CO = codon-optimized.

Study design

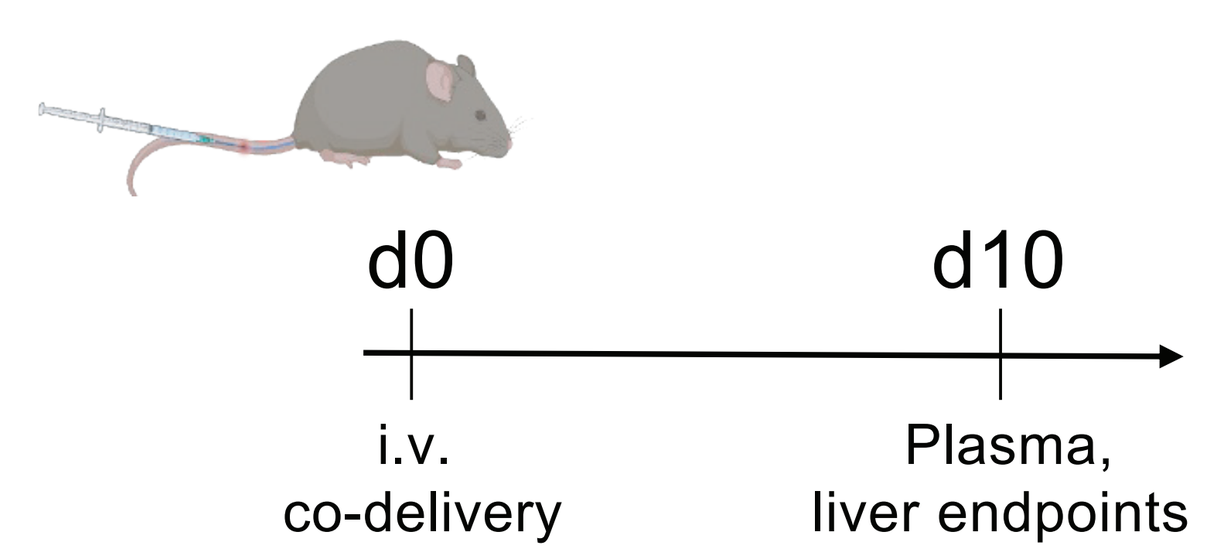
d0
d10
i.v.
co-delivery
Plasma,
liver endpoints

LNP: LP01, 0.5 mpk
AAV8: 0.3, 1.0, 3.0 e13 vg/kg

Liver endpoints: Vector copy number, % indels, % integration, and hATIII mRNA by ddPCR.
Plasma endpoint: hATIII expression by ELISA.
Sham controls
1a = irrelevant AAV,
1b = irrelevant LNP.

Vector copy numbers, indels, and integration in liver

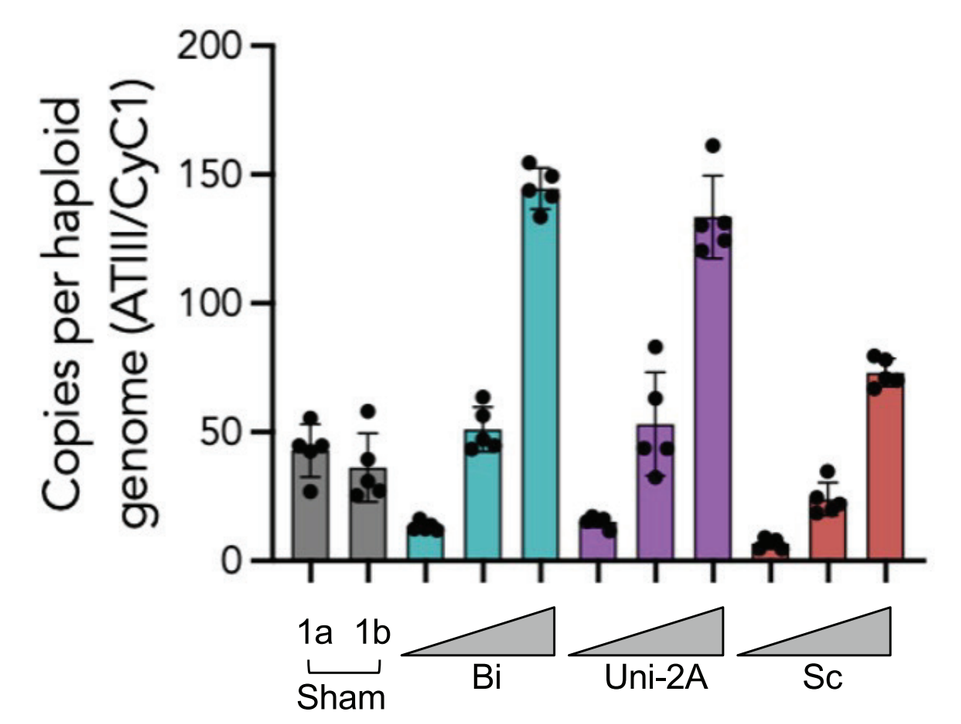
Copies per haploid genome (ATIII/CyC1)
1a 1b
Sham
Bi
Uni-2A
Sc

Result: Vector copy numbers in liver increase as a function of AAV dose.

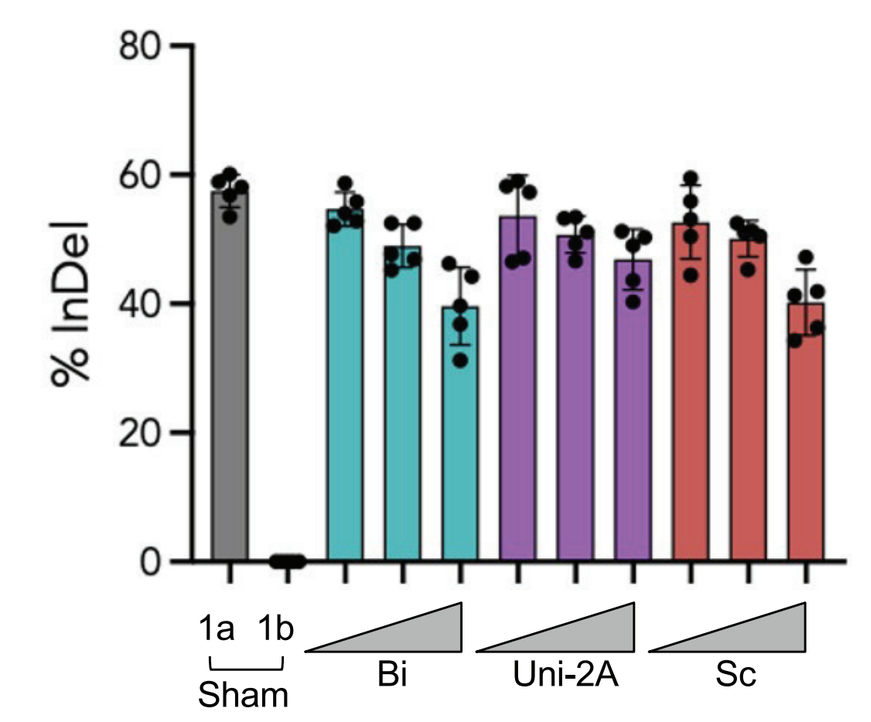
% InDel
1a 1b
Sham
Bi
Uni-2A
Sc

Result: Saturating editing was seen at the albumin locus, with AAV dose-dependent decreases due to cargo insertion.

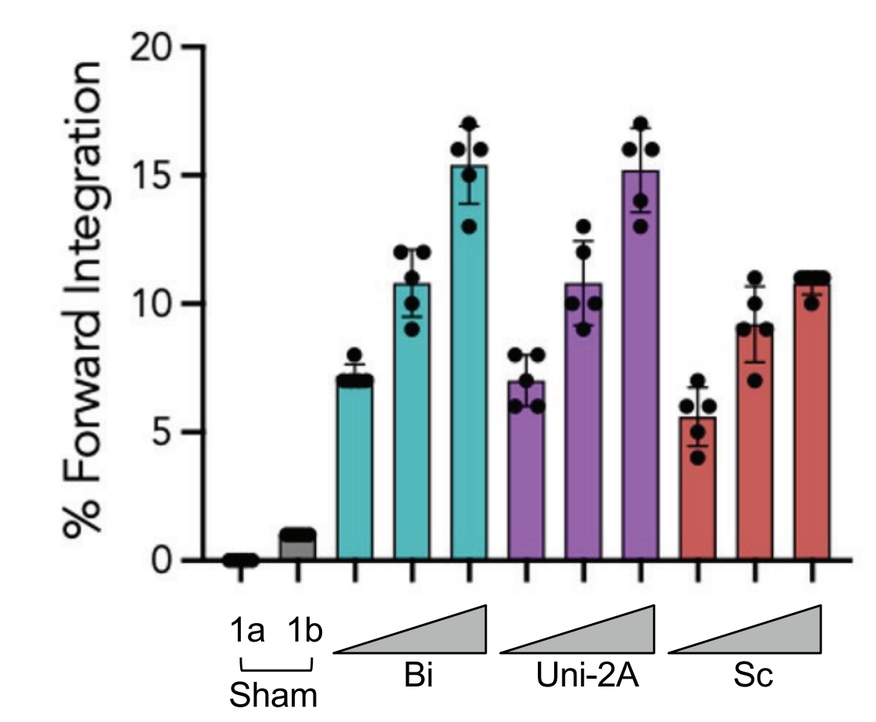
% Forward Integration
1a 1b
Sham
Bi
Uni-2A
Sc

Result: Up to 15% forward integration was detected for Bi and Uni-2A templates, with a maximum of 11% for the Sc template.

hATIII expression in liver and in plasma

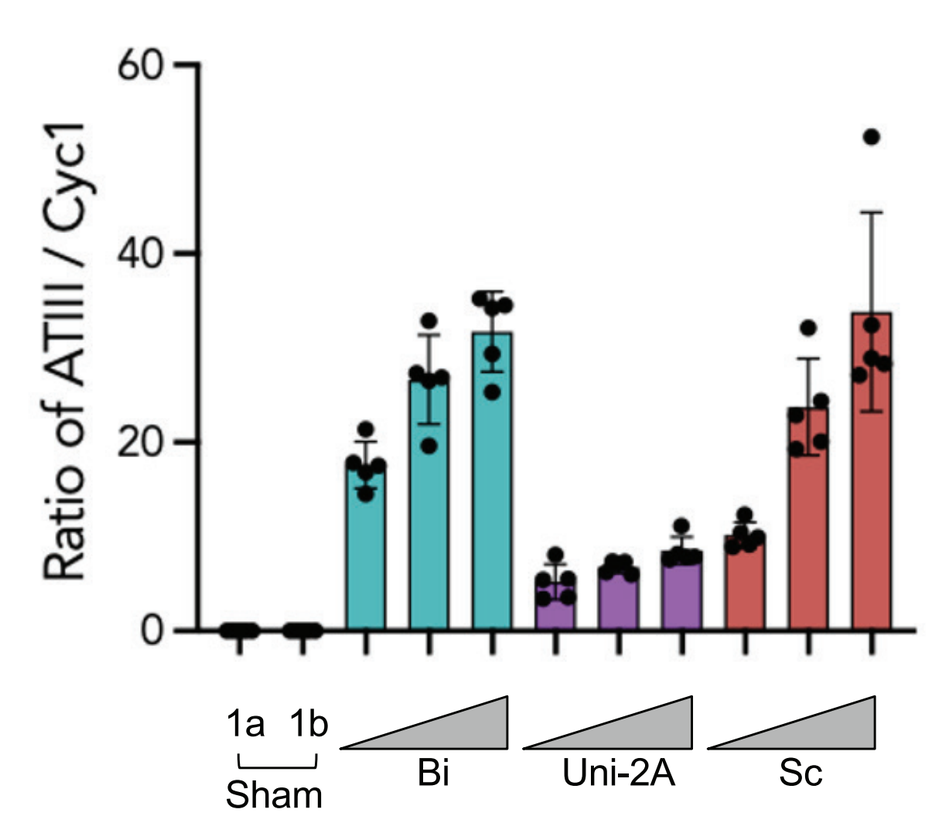
Ratio of ATIII / Cyc1
1a 1b
Sham
Bi
Uni-2A
Sc

Result: hATIII mRNA production in liver is highest with Bi and Sc templates.

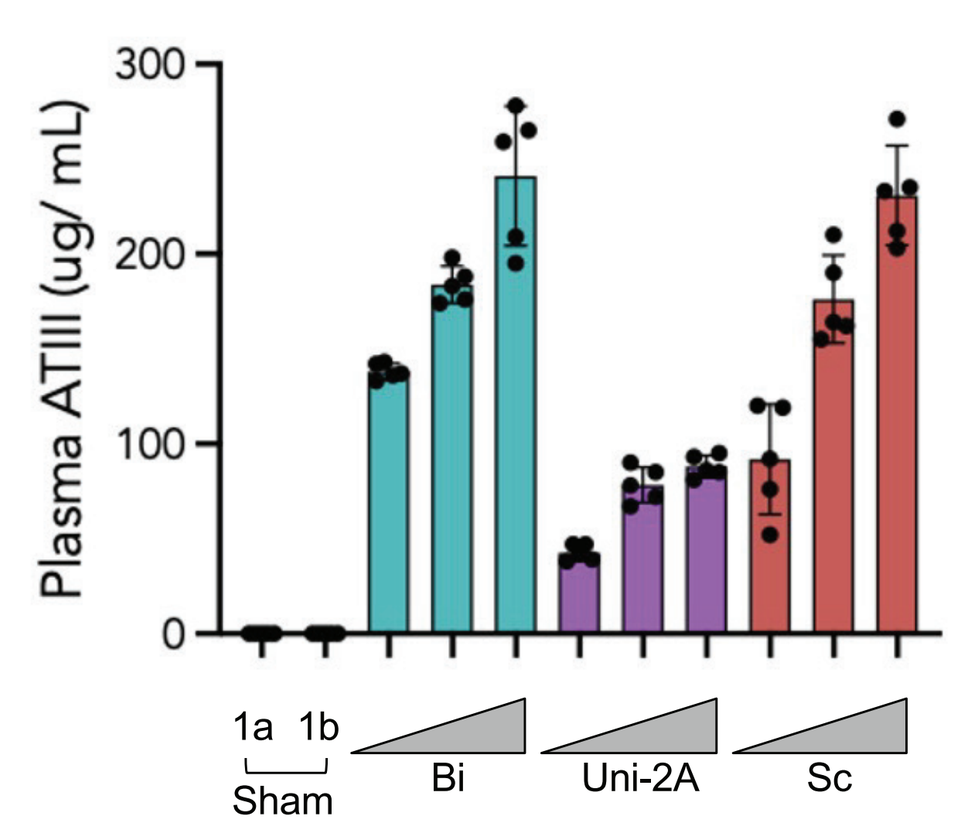
Plasma ATIII (ug/ mL)
1a 1b
Sham
Bi
Uni-2A
Sc

Result: AAV dose-dependent increases in hATIII in plasma were observed with a maximum of 240 ug/mL.

The scAAV format produces the most protein per unit integration

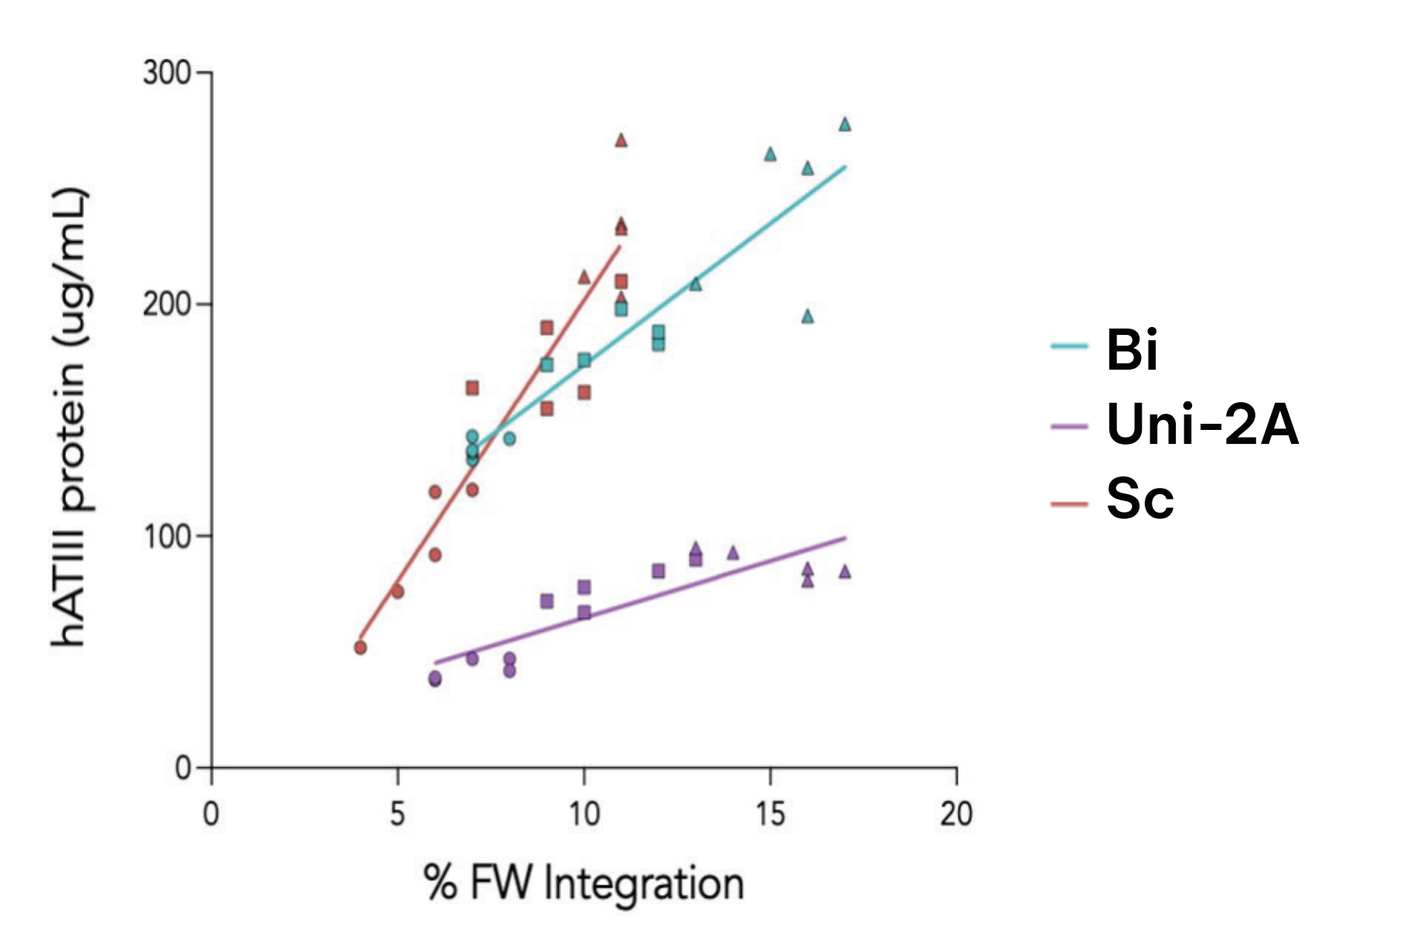
hATIII protein (ug/mL)
% FW Integration
Bi
Uni-2A
Sc

Result: Across dosing groups, the highest correlation between hATIII plasma protein and % integration was observed for the scAAV template.

Preclinical proof-of-concept study in non-human primates

Study design

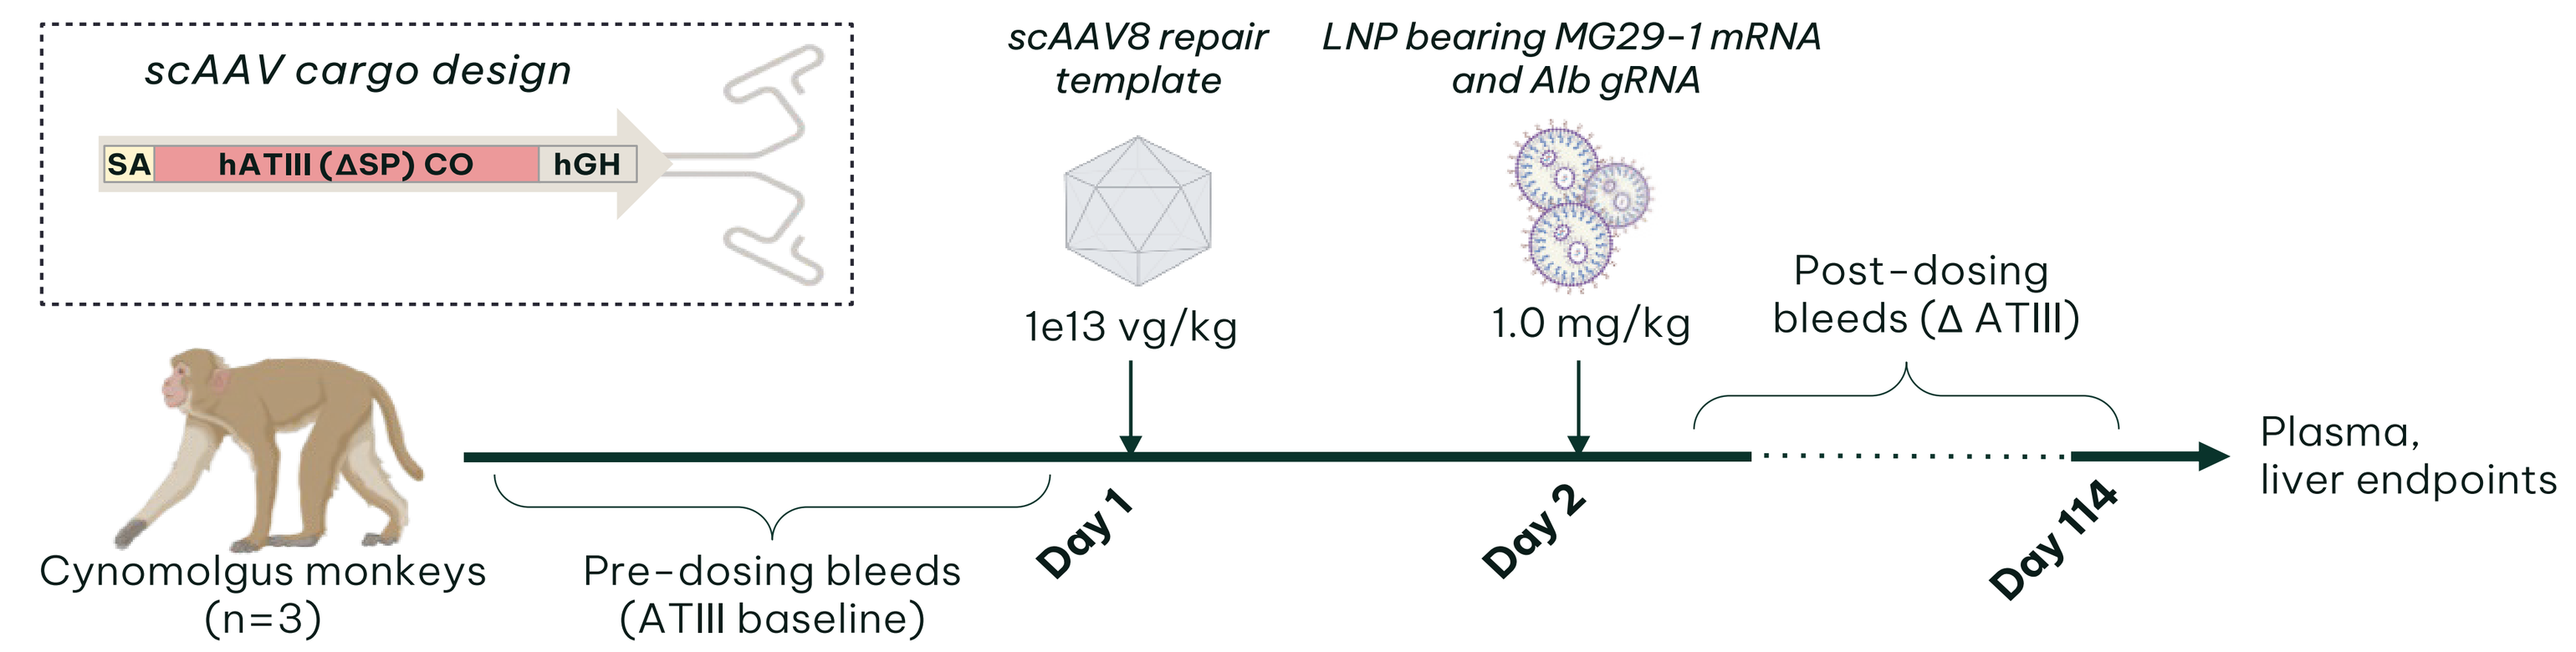
scAAV cargo design
SA hATIII (ΔSP) CO hGH
scAAV8 repair template
1e13 vg/kg
LNP bearing MG29-1 mRNA and Alb gRNA
1.0 mg/kg
Post-dosing bleeds (Δ ATIII)
Cynomolgus monkeys (n=3)
Pre-dosing bleeds (ATIII baseline)
Day 1
Day 2
Day 114
Plasma, liver endpoints

Dexamethasone (2 mg/kg) was administered i.v. at least 1 hr prior to LNP dosing, and methylprednisolone sodium succinate (5 mg/kg) was given i.m. at 6 and 24 hrs post-LNP. Blood was collected before and after dosing at regular intervals to measure plasma ATIII as well as other readouts.

Sustained plasma ATIII levels are observed

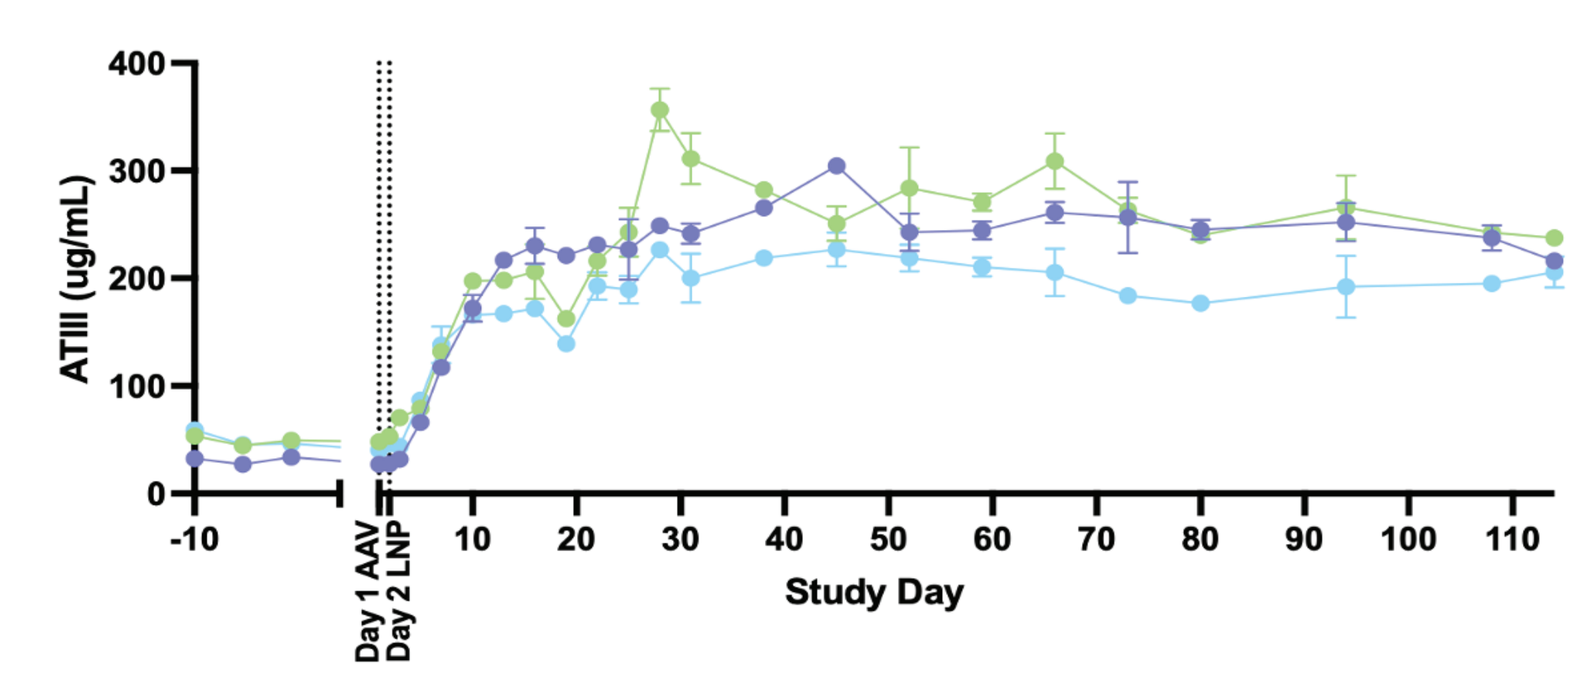
ATIII (ug/mL)
Day 1 AAV
Day 2 LNP
Study Day

Result: Plasma ATIII expression increases after dosing, reaching a plateau around study day 13. These levels are sustained through the end of the study on day 114.

Therapeutic levels of ATIII are achievable with the platform

Normal human plasma ATIII: ~150 ug/mL[3]

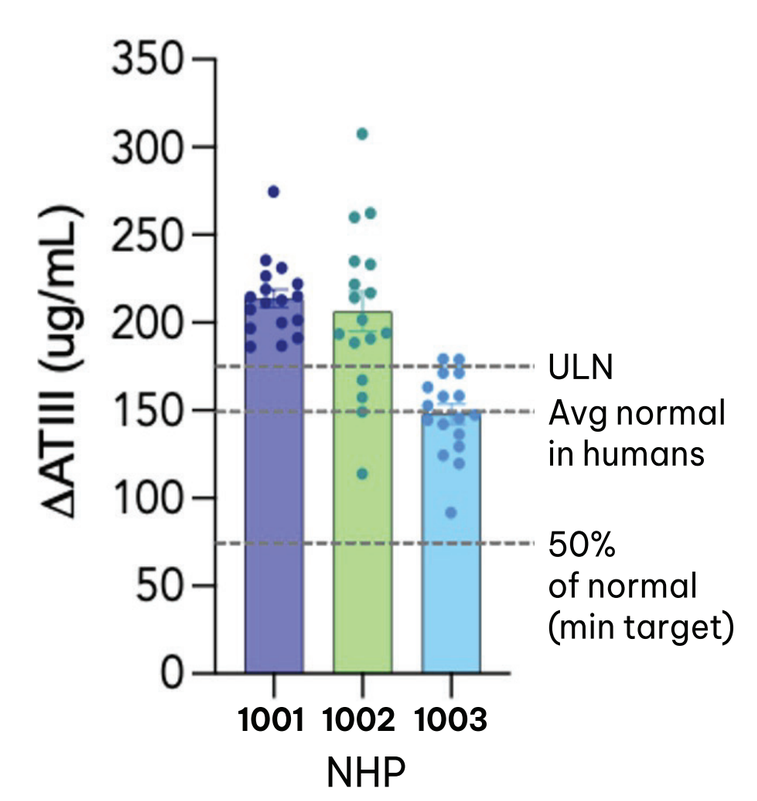
ΔATIII (ug/mL)
ULN
Avg normal in humans
50% of normal (min target)
1001 1002 1003
NHP

Results:

- The graph shows plasma ATIII levels on study days 13-114 after subtraction of the mean pre-dose level.
- Assuming normal human ATIII levels of ~150 ug/mL[3], the levels corresponding to the upper limit of normal (ULN), average, and 50% of normal are indicated.
- The levels achieved in this study are sufficient to treat severe thrombophilia, defined as 40-60% of normal.

Plasma ATIII is biochemically active

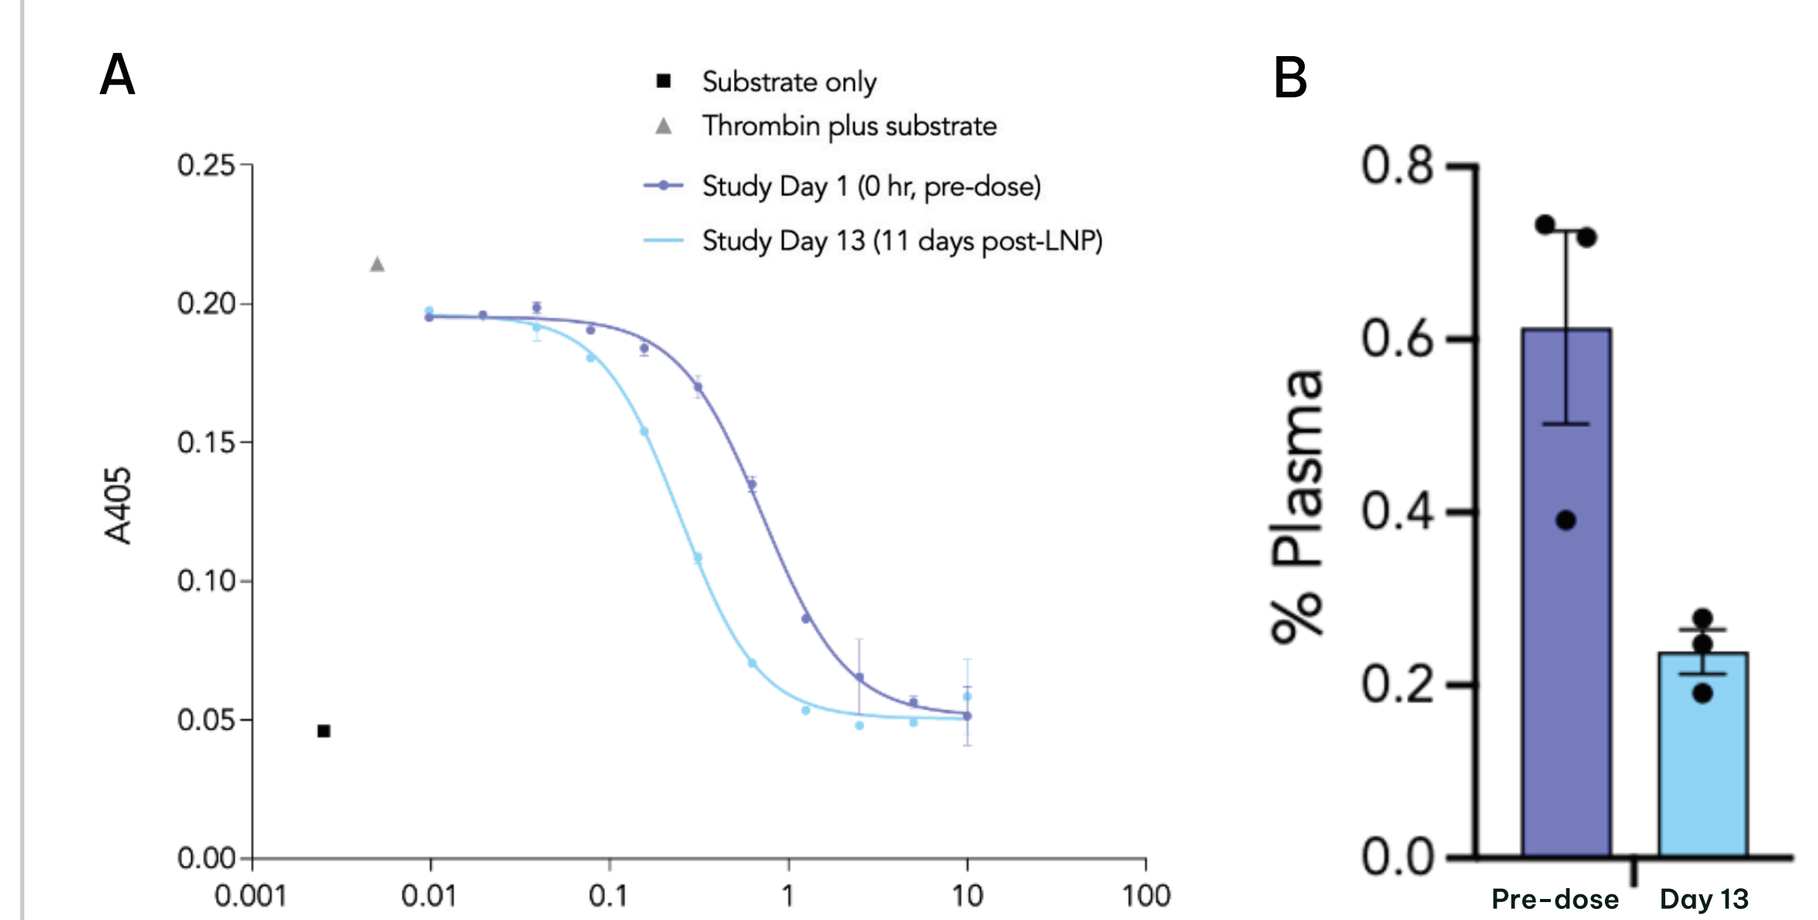
A
Substrate only
Thrombin plus substrate
Study Day 1 (0 hr, pre-dose)
Study Day 13 (11 days post-LNP)
A405
Plasma (%)
B
% Plasma
Pre-dose
Day 13

Result: *In vitro* thrombin inhibition (BPS Bioscience) was used to infer the activity of the expressed ATIII.
A) Post-dose (day 13) plasma more potently inhibited thrombin activity relative to pre-dose plasma (one representative animal shown).
B) IC50 percent plasma concentration across all three animals shows greater biochemical potency post-dose.

ATIII expression has minimal effect on hemostasis readouts

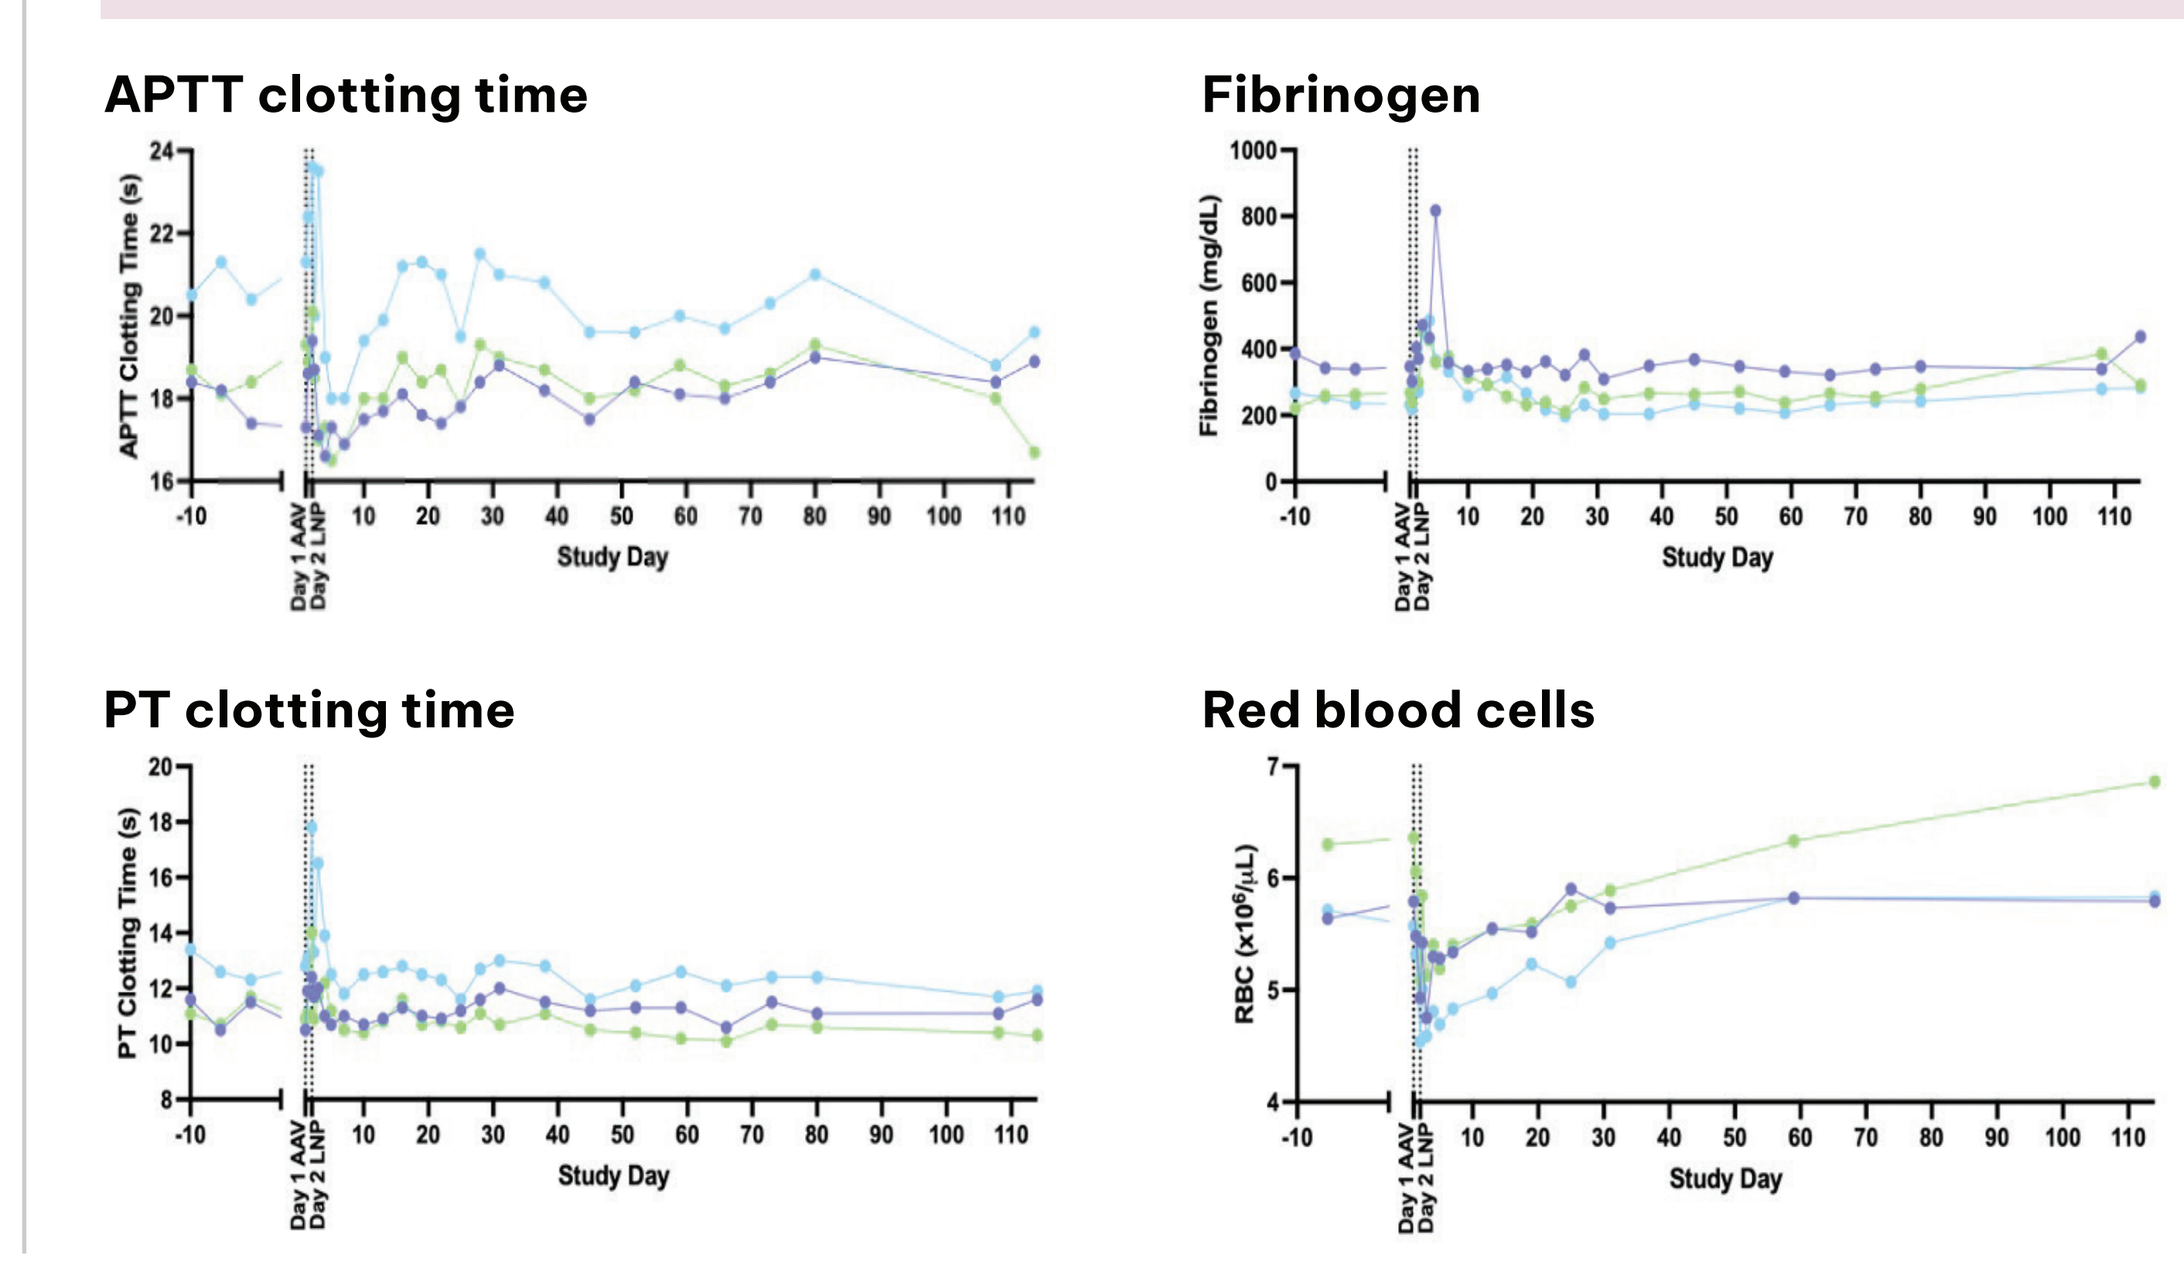
APTT clotting time
Fibrinogen
PT clotting time
Red blood cells
APTT Clotting Time (s)
Fibrinogen (mg/dL)
PT Clotting Time (s)
Study Day

Result: Plasma readouts for activated partial thromboplastin time (APTT), prothrombin time (PT), fibrinogen, and red blood cell counts (RBC) showed no significant changes.

Conclusions

- ATIII deficiency is a clotting disorder that, while currently manageable with DOACs, presents a significant treatment burden for patients facing surgery or pregnancy. Here we present a durable, "one-and-done" targeted gene integration approach potentially capable of lifelong restoration of normal ATIII levels.
- The most efficient template in terms of protein production per unit integration was scAAV.
- In NHPs, the scAAV format produced sustained, therapeutically relevant levels of biochemically active ATIII with minimal safety findings.
 Off-target and off-tissue editing by the nuclease and guide is minimal as assessed as part of our lead hemophilia A program (Poster PB2040).
- By using the same molecular approach established by our hemophilia A program, we demonstrate the potential to address multiple secreted protein disorders with the same platform.

References

1. Corral J, Morena-Barrio ME, Vicente V (2018).
 The genetics of antithrombin. *Thromb. Res. 169, 23-29.*
2. White K and Hunt BJ (2022). Inherited antithrombin deficiency in pregnancy. *Thrombosis Update 6, 100094.*
3. Conard J, Brosstad F, Lie Larsen M, Samama M, Abildgaard U (1983).
 Molar antithrombin concentration in normal human plasma. *Haemostasis 13, 363–8*

Contact information

Alan R. Brooks (alan.brooks@metagenomi.co)
Katalin Kauser (katalin.kauser@metagenomi.co)

5959 Horton St,
Emeryville,
California 94608
+1 (510) 871-4880
www.metagenomi.co